HER2-XPAT and EGFR-XPAT: Pro-Drug T Cell Engagers (TCEs) Engineered to Address *On*-Target, *Off*-Tumor Toxicity With Potent Efficacy *in vitro* and *in vivo* and Large Safety Margins in NHP

Fiore Cattaruzza, Ayesha Nazeer, Zachary Lange, Mikhail Hammond, Caitlin Koski, Trang Dao-Pick, Angela Henkensiefken, Mika K. Derynck, Bryan A. Irving, and Volker Schellenberger

Amunix Pharmaceuticals, Inc. Mountain View, CA

INTRODUCTION

Bispecific T Cell Engagers (TCEs) are effective at inducing remissions in hematologic cancers, but their use in solid tumors has been challenging due to their extreme potency and unforgiving toxicity against target expression in healthy tissue. To address this challenge, Amunix has developed conditionally active TCEs, XPATs or **X**TENylated **P**rotease-**A**ctivated bispecific **T** Cell Engagers targeting HER2 and EGFR that exploit the dysregulated protease activity present in tumors vs. healthy tissues, enabling expansion of the therapeutic index. The XPAT core consists of 2 single chain antibody fragments (scFvs) targeting CD3 and the tumor target. Two unstructured polypeptide masks (XTEN) are attached to the core that sterically reduce target engagement and extend protein half-life. Protease cleavage sites at the base of the XTEN masks enable proteolytic activation of XPAT in the tumor microenvironment, unleashing a small, highly potent TCE. In healthy tissues, where protease activity is tightly regulated, XPATs should remain predominantly inactive as intact prodrugs. In addition to localized activation, the short half-life of the unmasked PAT form should further widen the therapeutic index while providing the potency of T-cell immunity to improve the eradication of solid tumors.

XPAT PLATFORM

XPATs Are XTENylated Protease-Activated T Cell Engagers

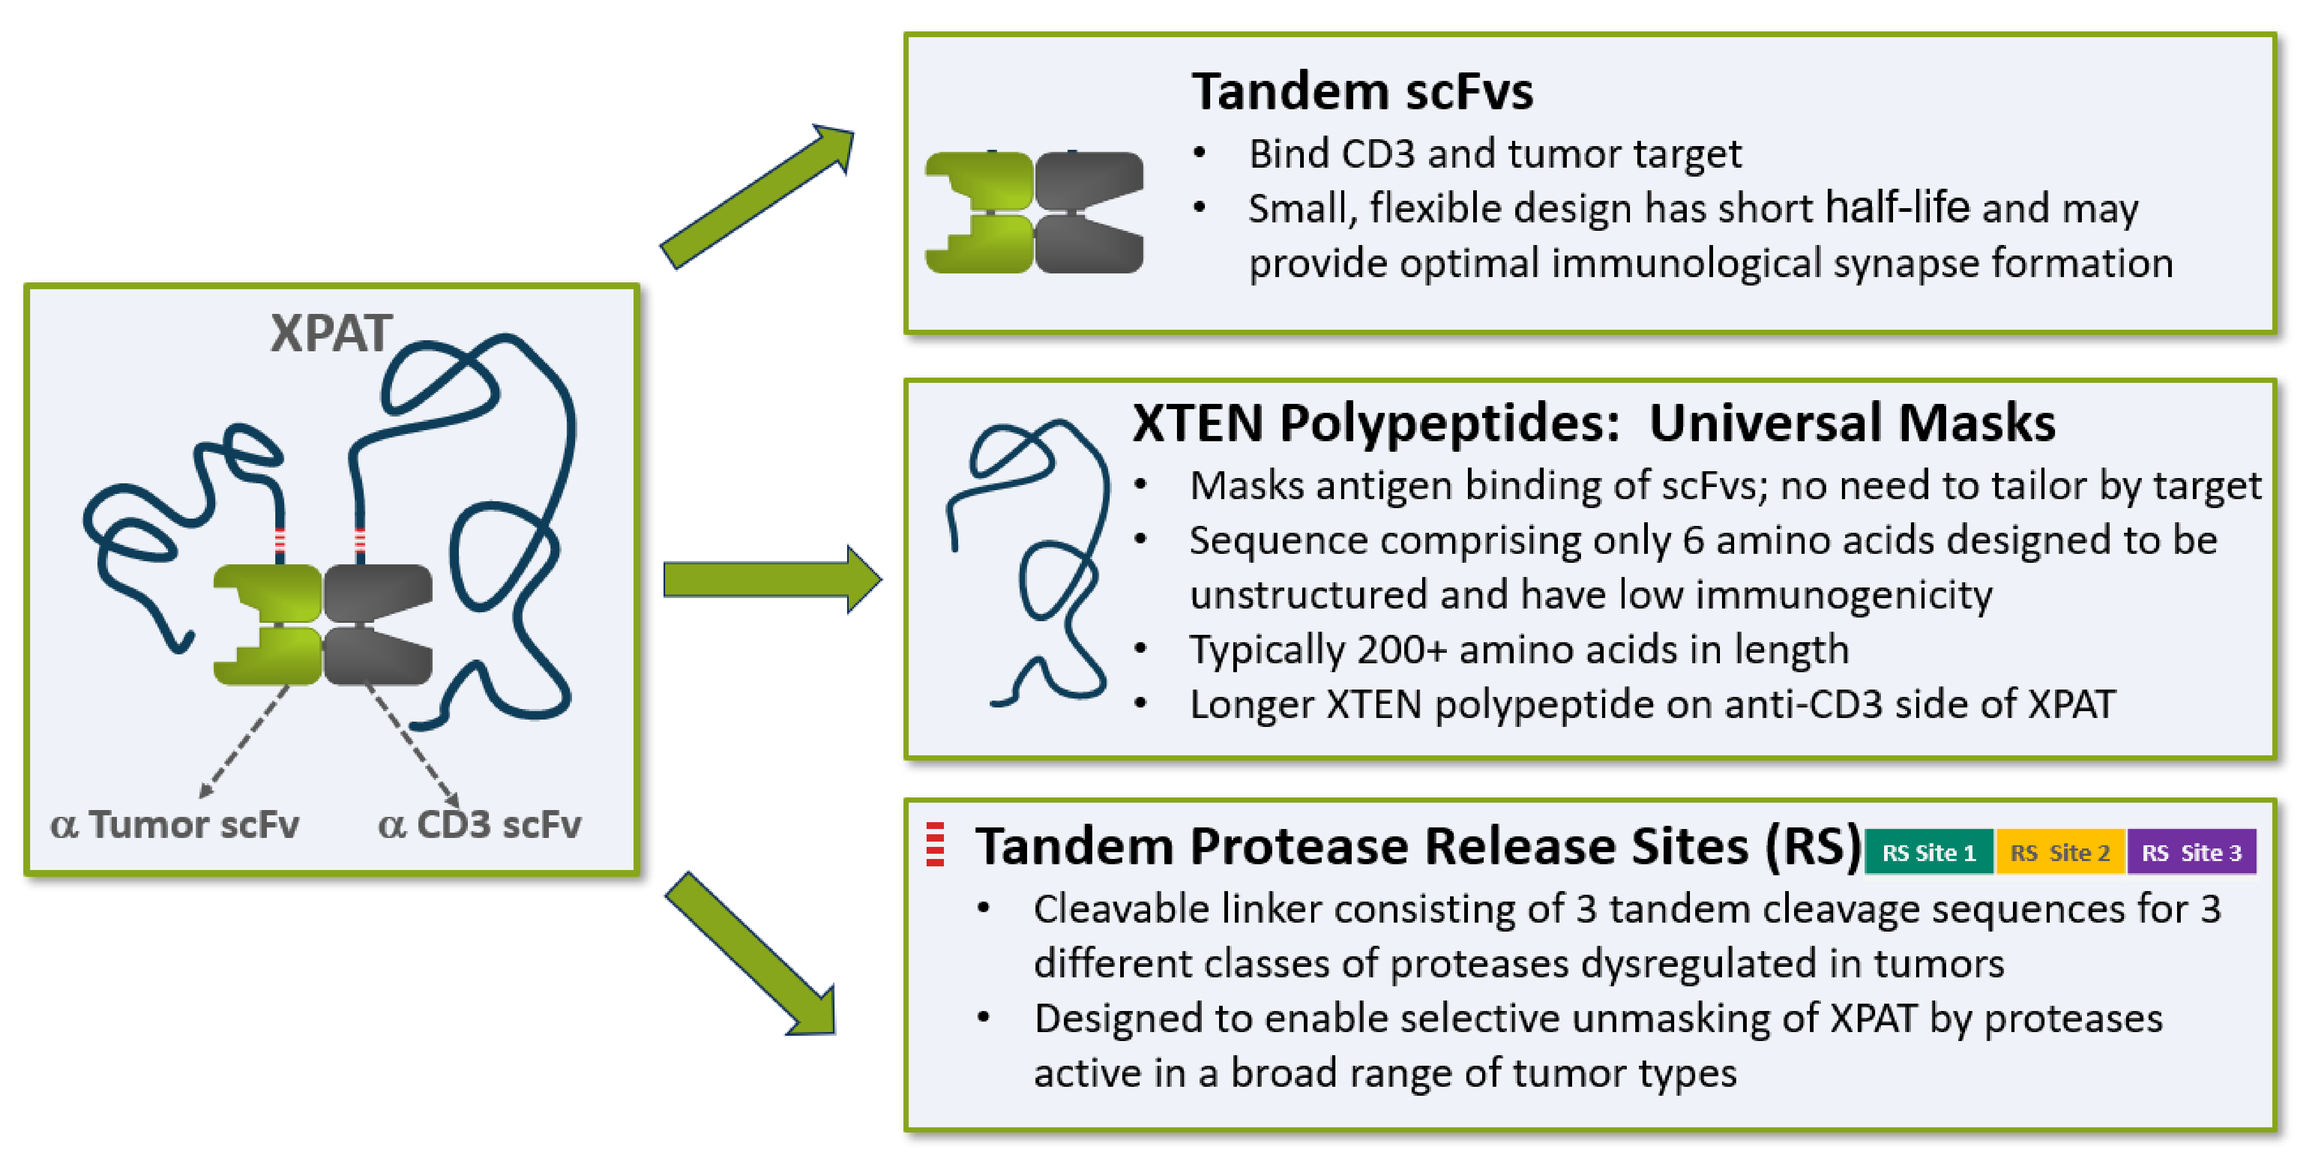
XPAT
α Tumor scFv
α CD3 scFv
Tandem scFvs
Bind CD3 and tumor target
Small, flexible design has short half-life and may provide optimal immunological synapse formation
XTEN Polypeptides: Universal Masks
Masks antigen binding of scFvs; no need to tailor by target
Sequence comprising only 6 amino acids designed to be unstructured and have low immunogenicity
Typically 200+ amino acids in length
Longer XTEN polypeptide on anti-CD3 side of XPAT
Tandem Protease Release Sites (RS)
RS Site 1
RS Site 2
RS Site 3
Cleavable linker consisting of 3 tandem cleavage sequences for 3 different classes of proteases dysregulated in tumors
Designed to enable selective unmasking of XPAT by proteases active in a broad range of tumor types

XPATs Enable Localized Tumor Killing, Limiting Toxicity Against Healthy Tissue Expressing the Target Antigen

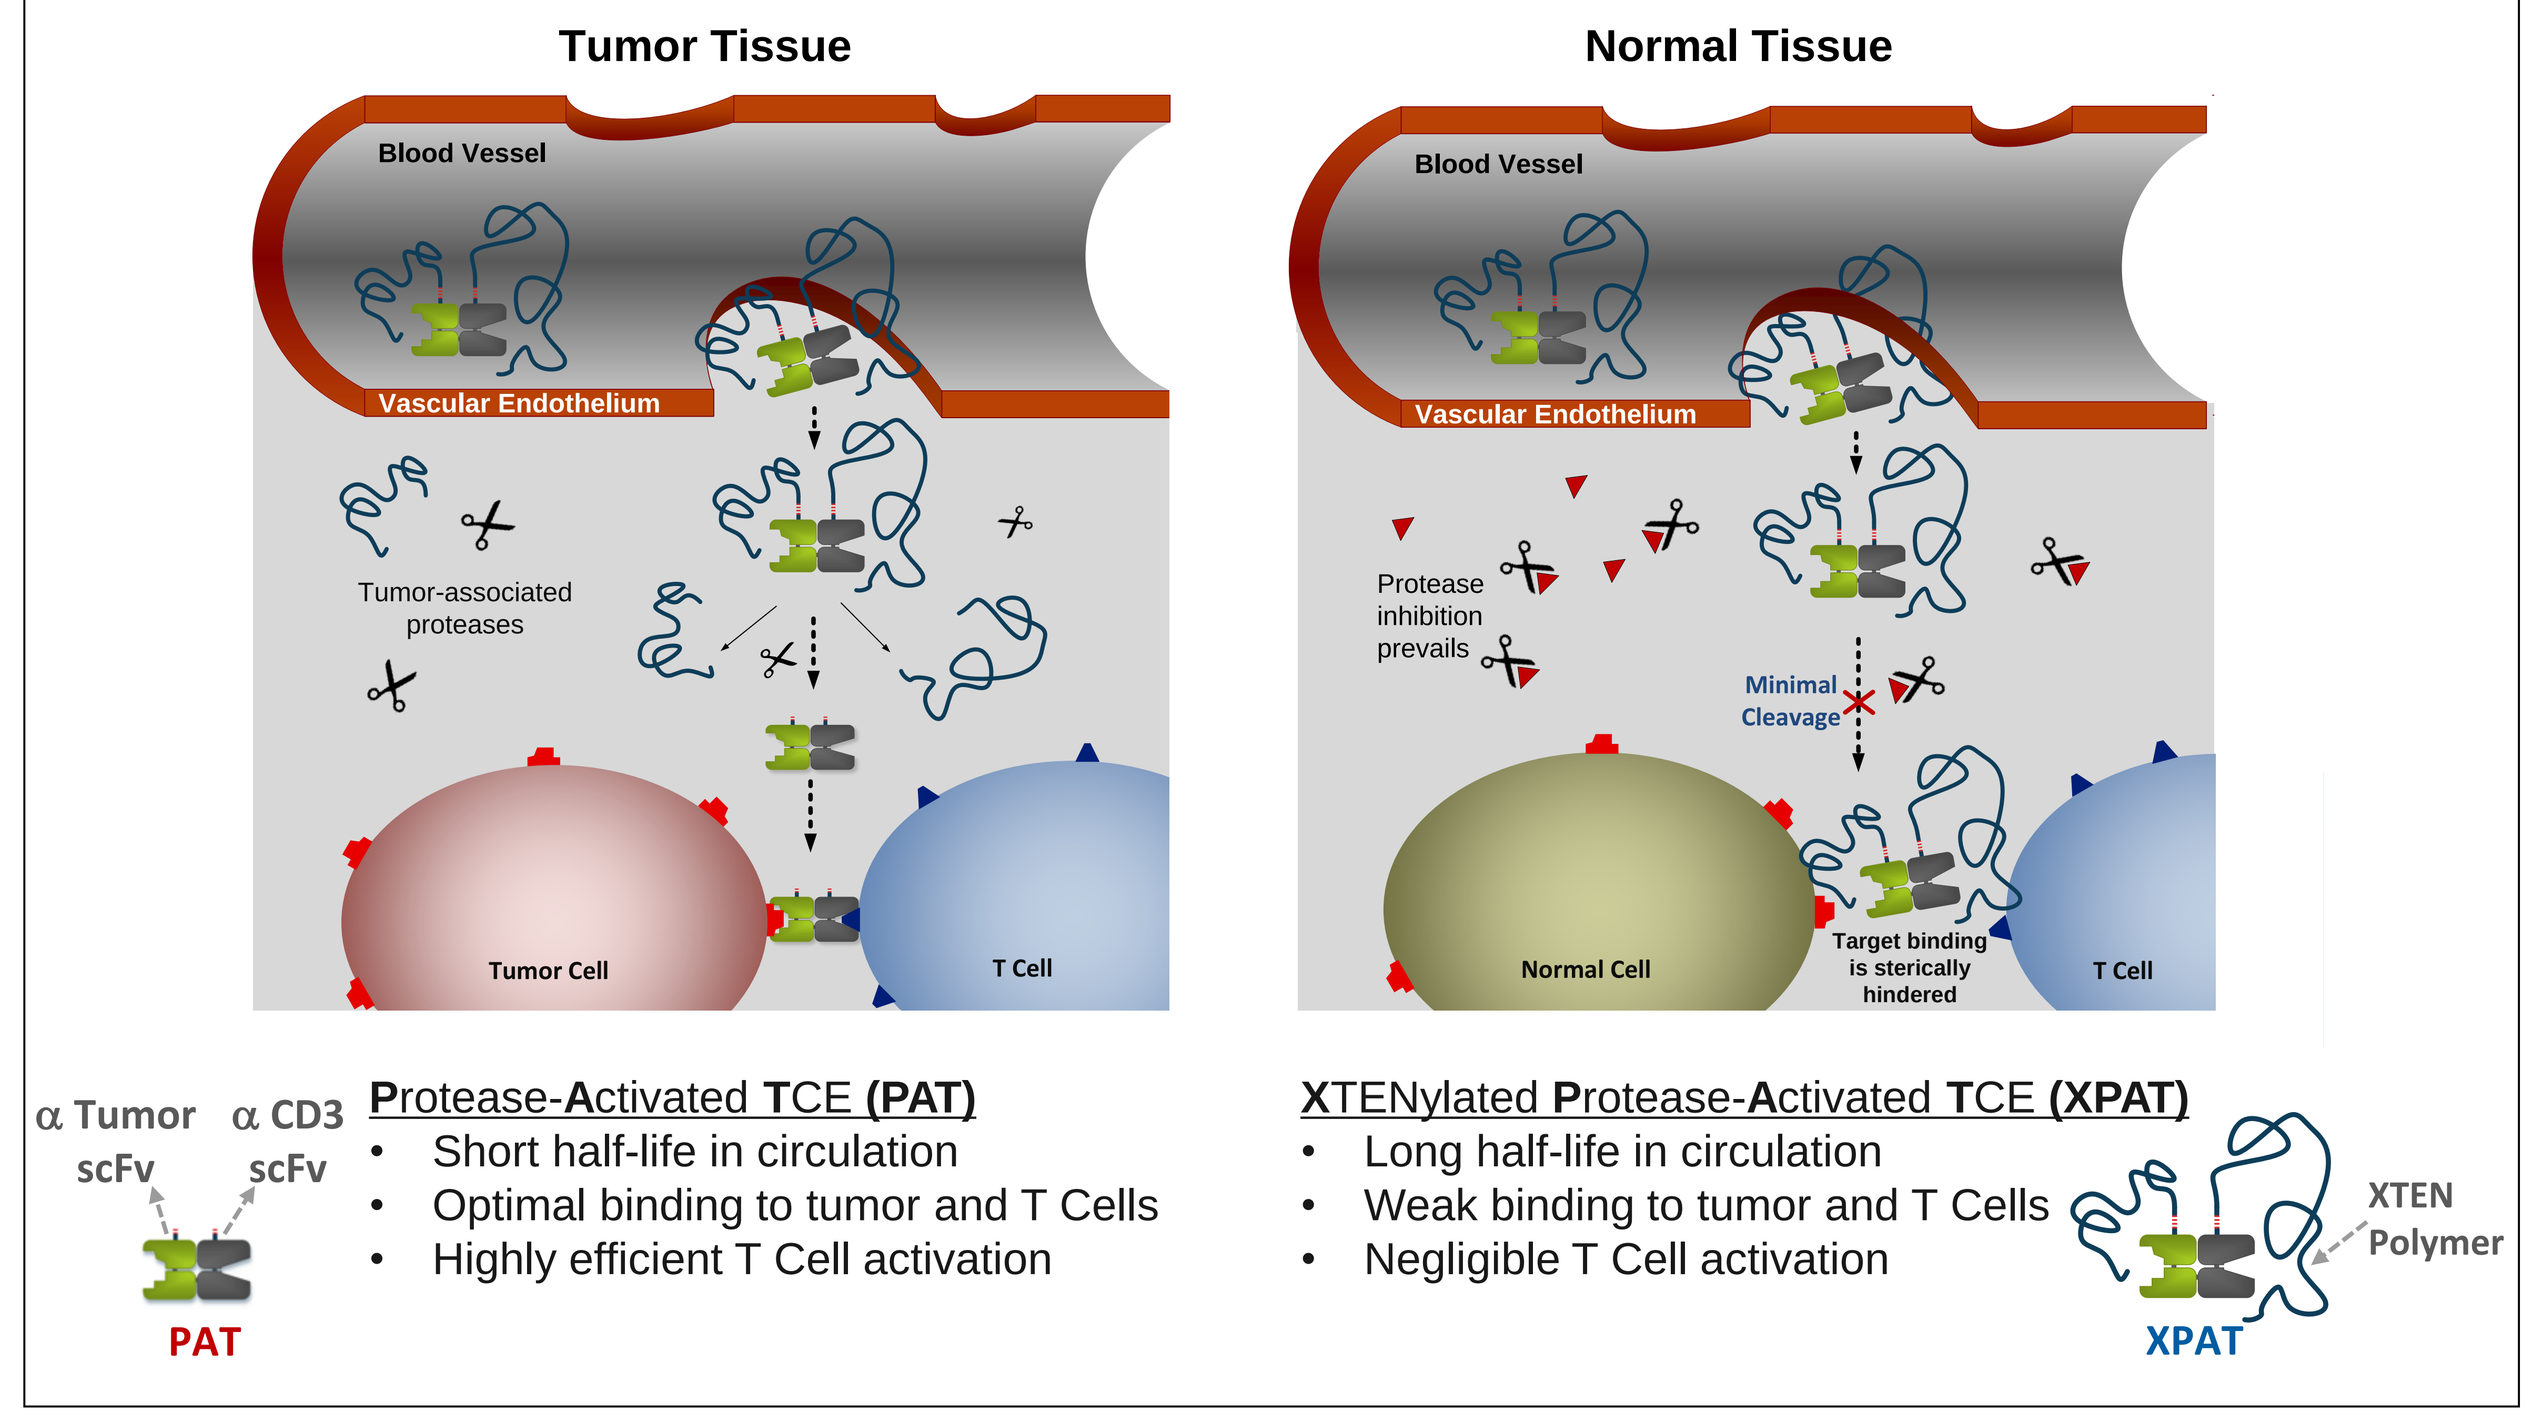
Tumor Tissue
Normal Tissue
Blood Vessel
Vascular Endothelium
Tumor-associated proteases
Tumor Cell
T Cell
Protease inhibition prevails
Minimal Cleavage
Normal Cell
Target binding is sterically hindered
α Tumor scFv
α CD3 scFv
PAT
Protease-Activated TCE (PAT)
Short half-life in circulation
Optimal binding to tumor and T Cells
Highly efficient T Cell activation
XTENylated Protease-Activated TCE (XPAT)
Long half-life in circulation
Weak binding to tumor and T Cells
Negligible T Cell activation
XTEN Polymer
XPAT

RESULTS

Figure 1. XTEN Polypeptide Masks on HER2-XPAT Significantly Reduce T Cell–Mediated Cytotoxicity and T Cell Activation *in vitro*

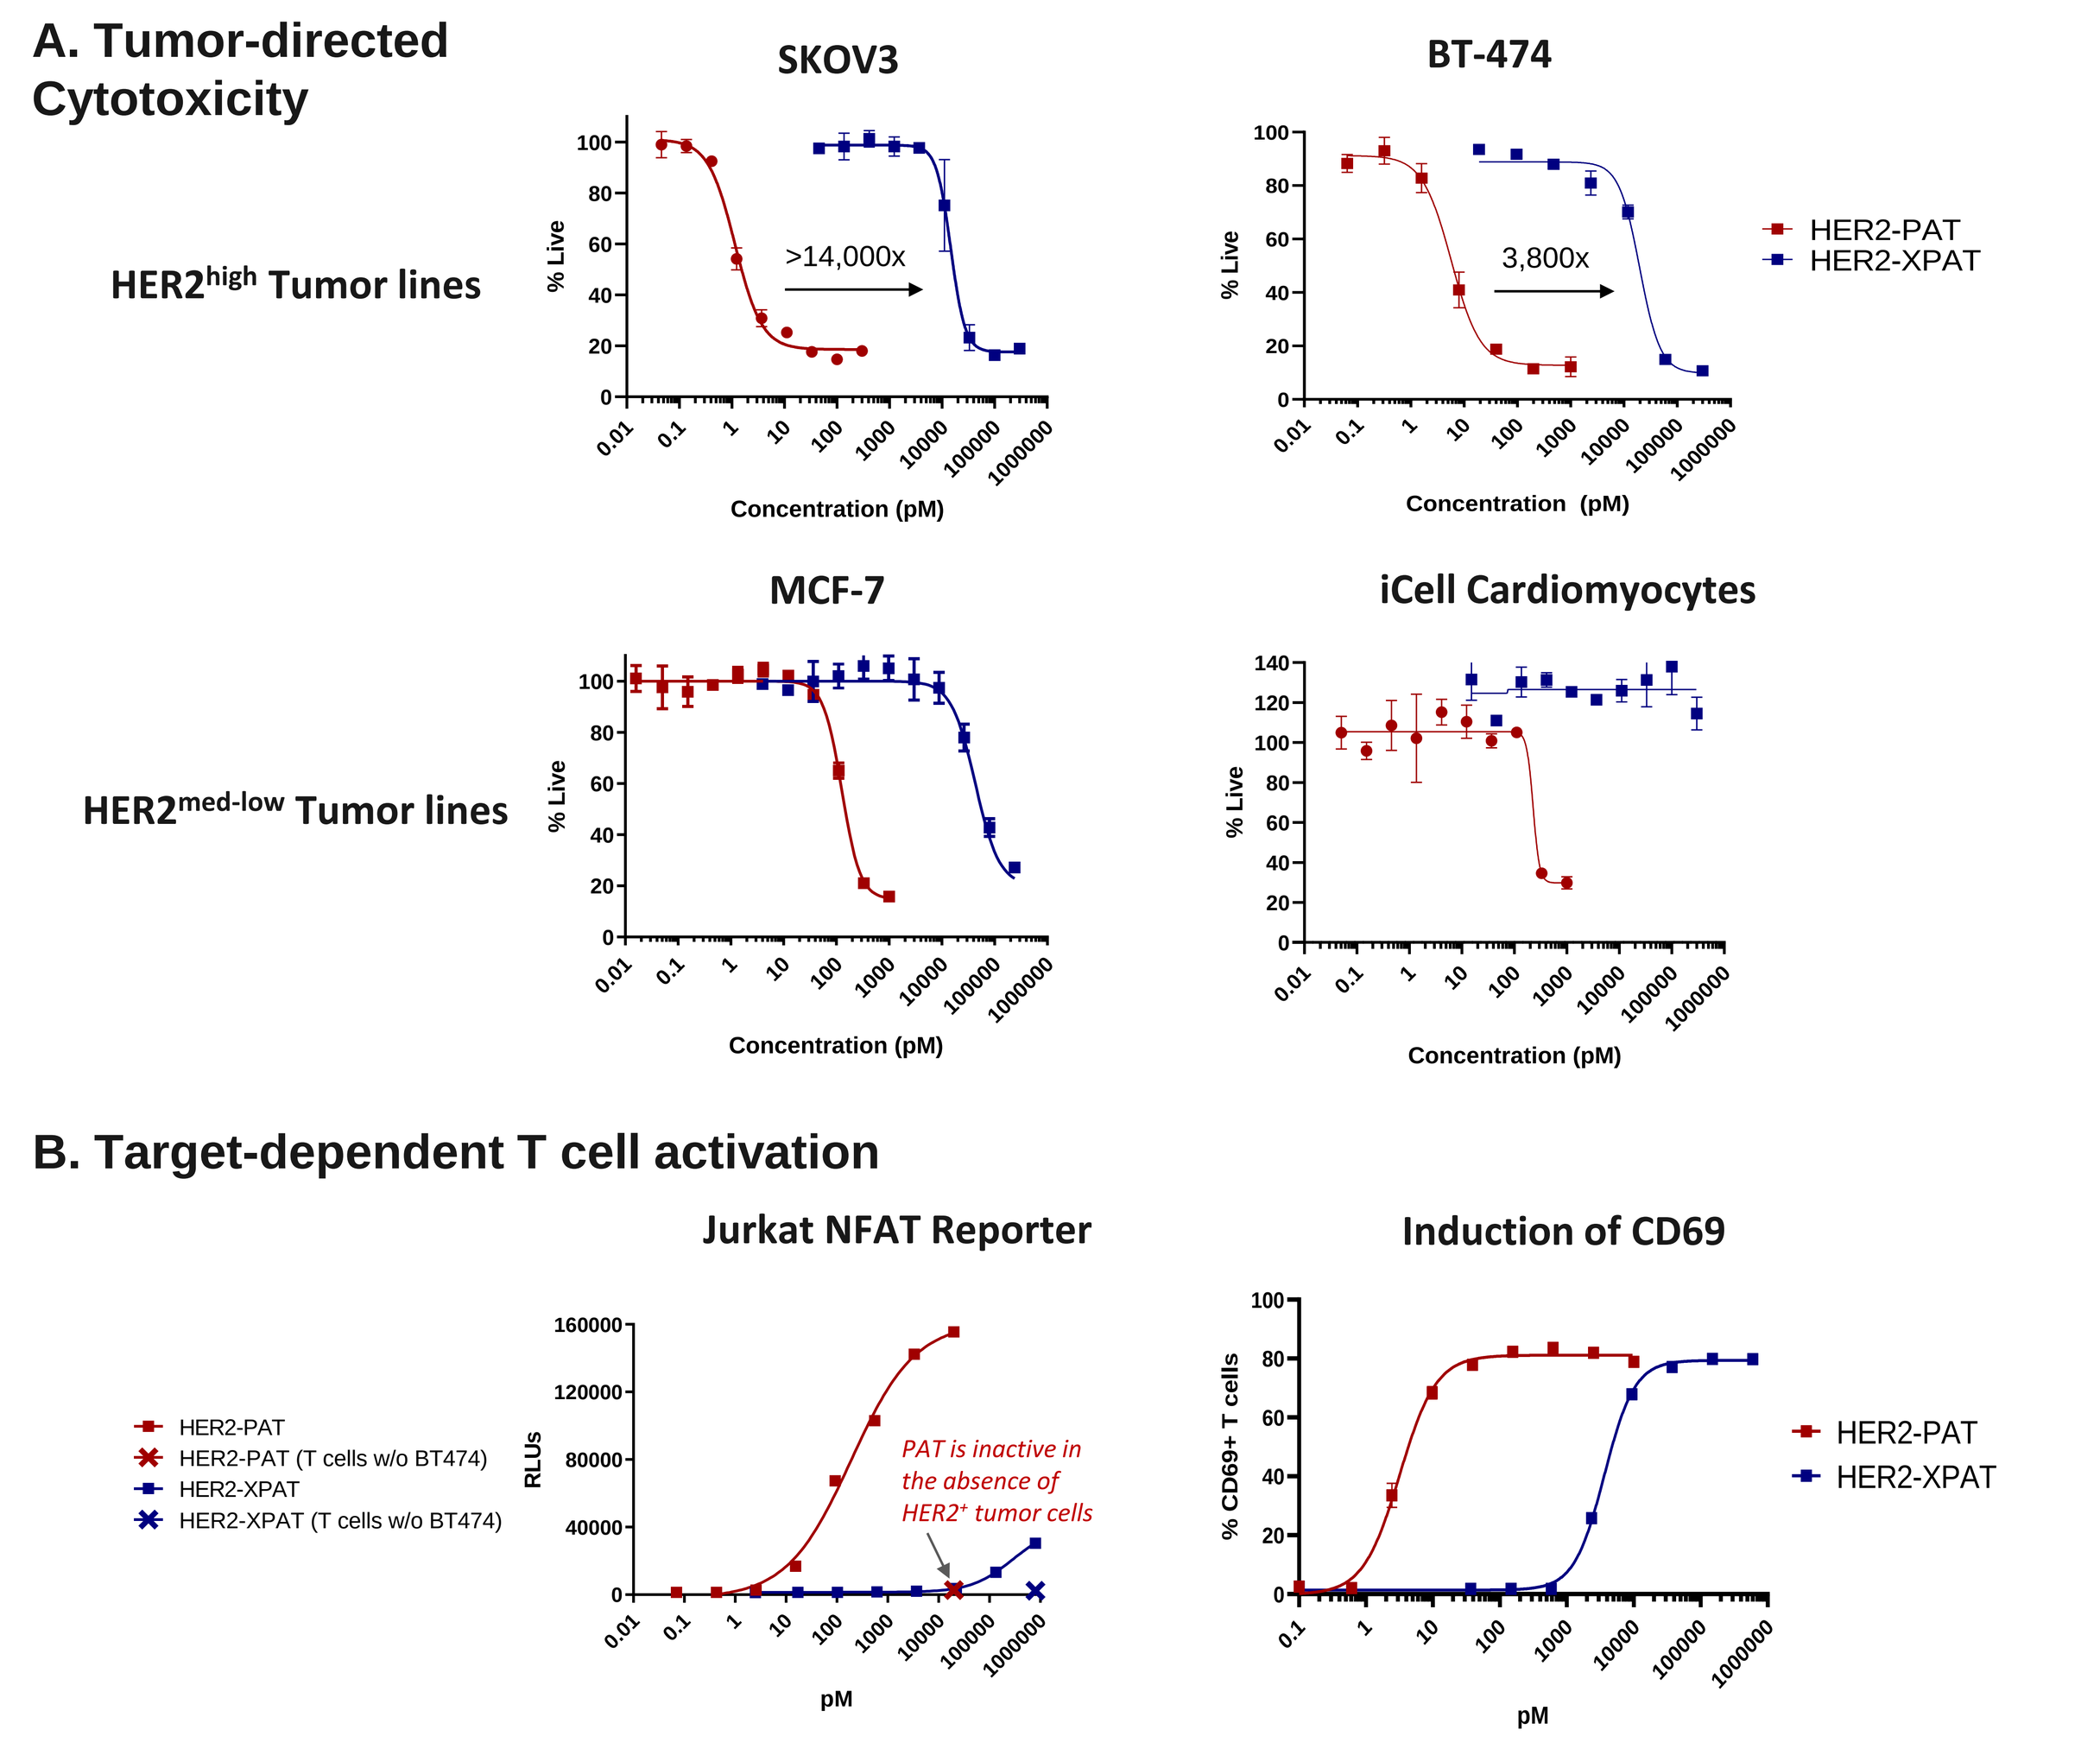
A. Tumor-directed Cytotoxicity
HER2high Tumor lines
HER2med-low Tumor lines
SKOV3
BT-474
MCF-7
iCell Cardiomyocytes
>14,000x
3,800x
% Live
Concentration (pM)
HER2-PAT
HER2-XPAT
B. Target-dependent T cell activation
Jurkat NFAT Reporter
Induction of CD69
HER2-PAT (T cells w/o BT474)
HER2-XPAT (T cells w/o BT474)
RLUs
% CD69+ T cells
PAT is inactive in the absence of HER2+ tumor cells
pM

A) Cytotoxicity was quantified using Cell Titre-Glo Luminescent Cell Viability Assay following a 48 hour incubation of huPBMCs and the indicated tumor targets or human iCell Cardiomyocytes at a 10:1 ratio (SKOV-3) or 5:1 ratio (others). Co-cultures were treated with HER2-XPAT or HER2-PAT at the concentrations shown. **B)** Jurkat reporter T cells were incubated with or without BT-474 cells at a 5:1 E:T ratio for 6 hours and NFAT-induced Luciferase activity quantified in response to the test articles. Surface CD69 expression was evaluated on T cells by flow cytometry following a 72 hour co-incubation of PBMCs and SKOV3 cells at a 5:1 E:T ratio with test articles at the indicated concentrations.

Figure *2.* HER2-XPAT Induces Robust Tumor Regressions in Mice

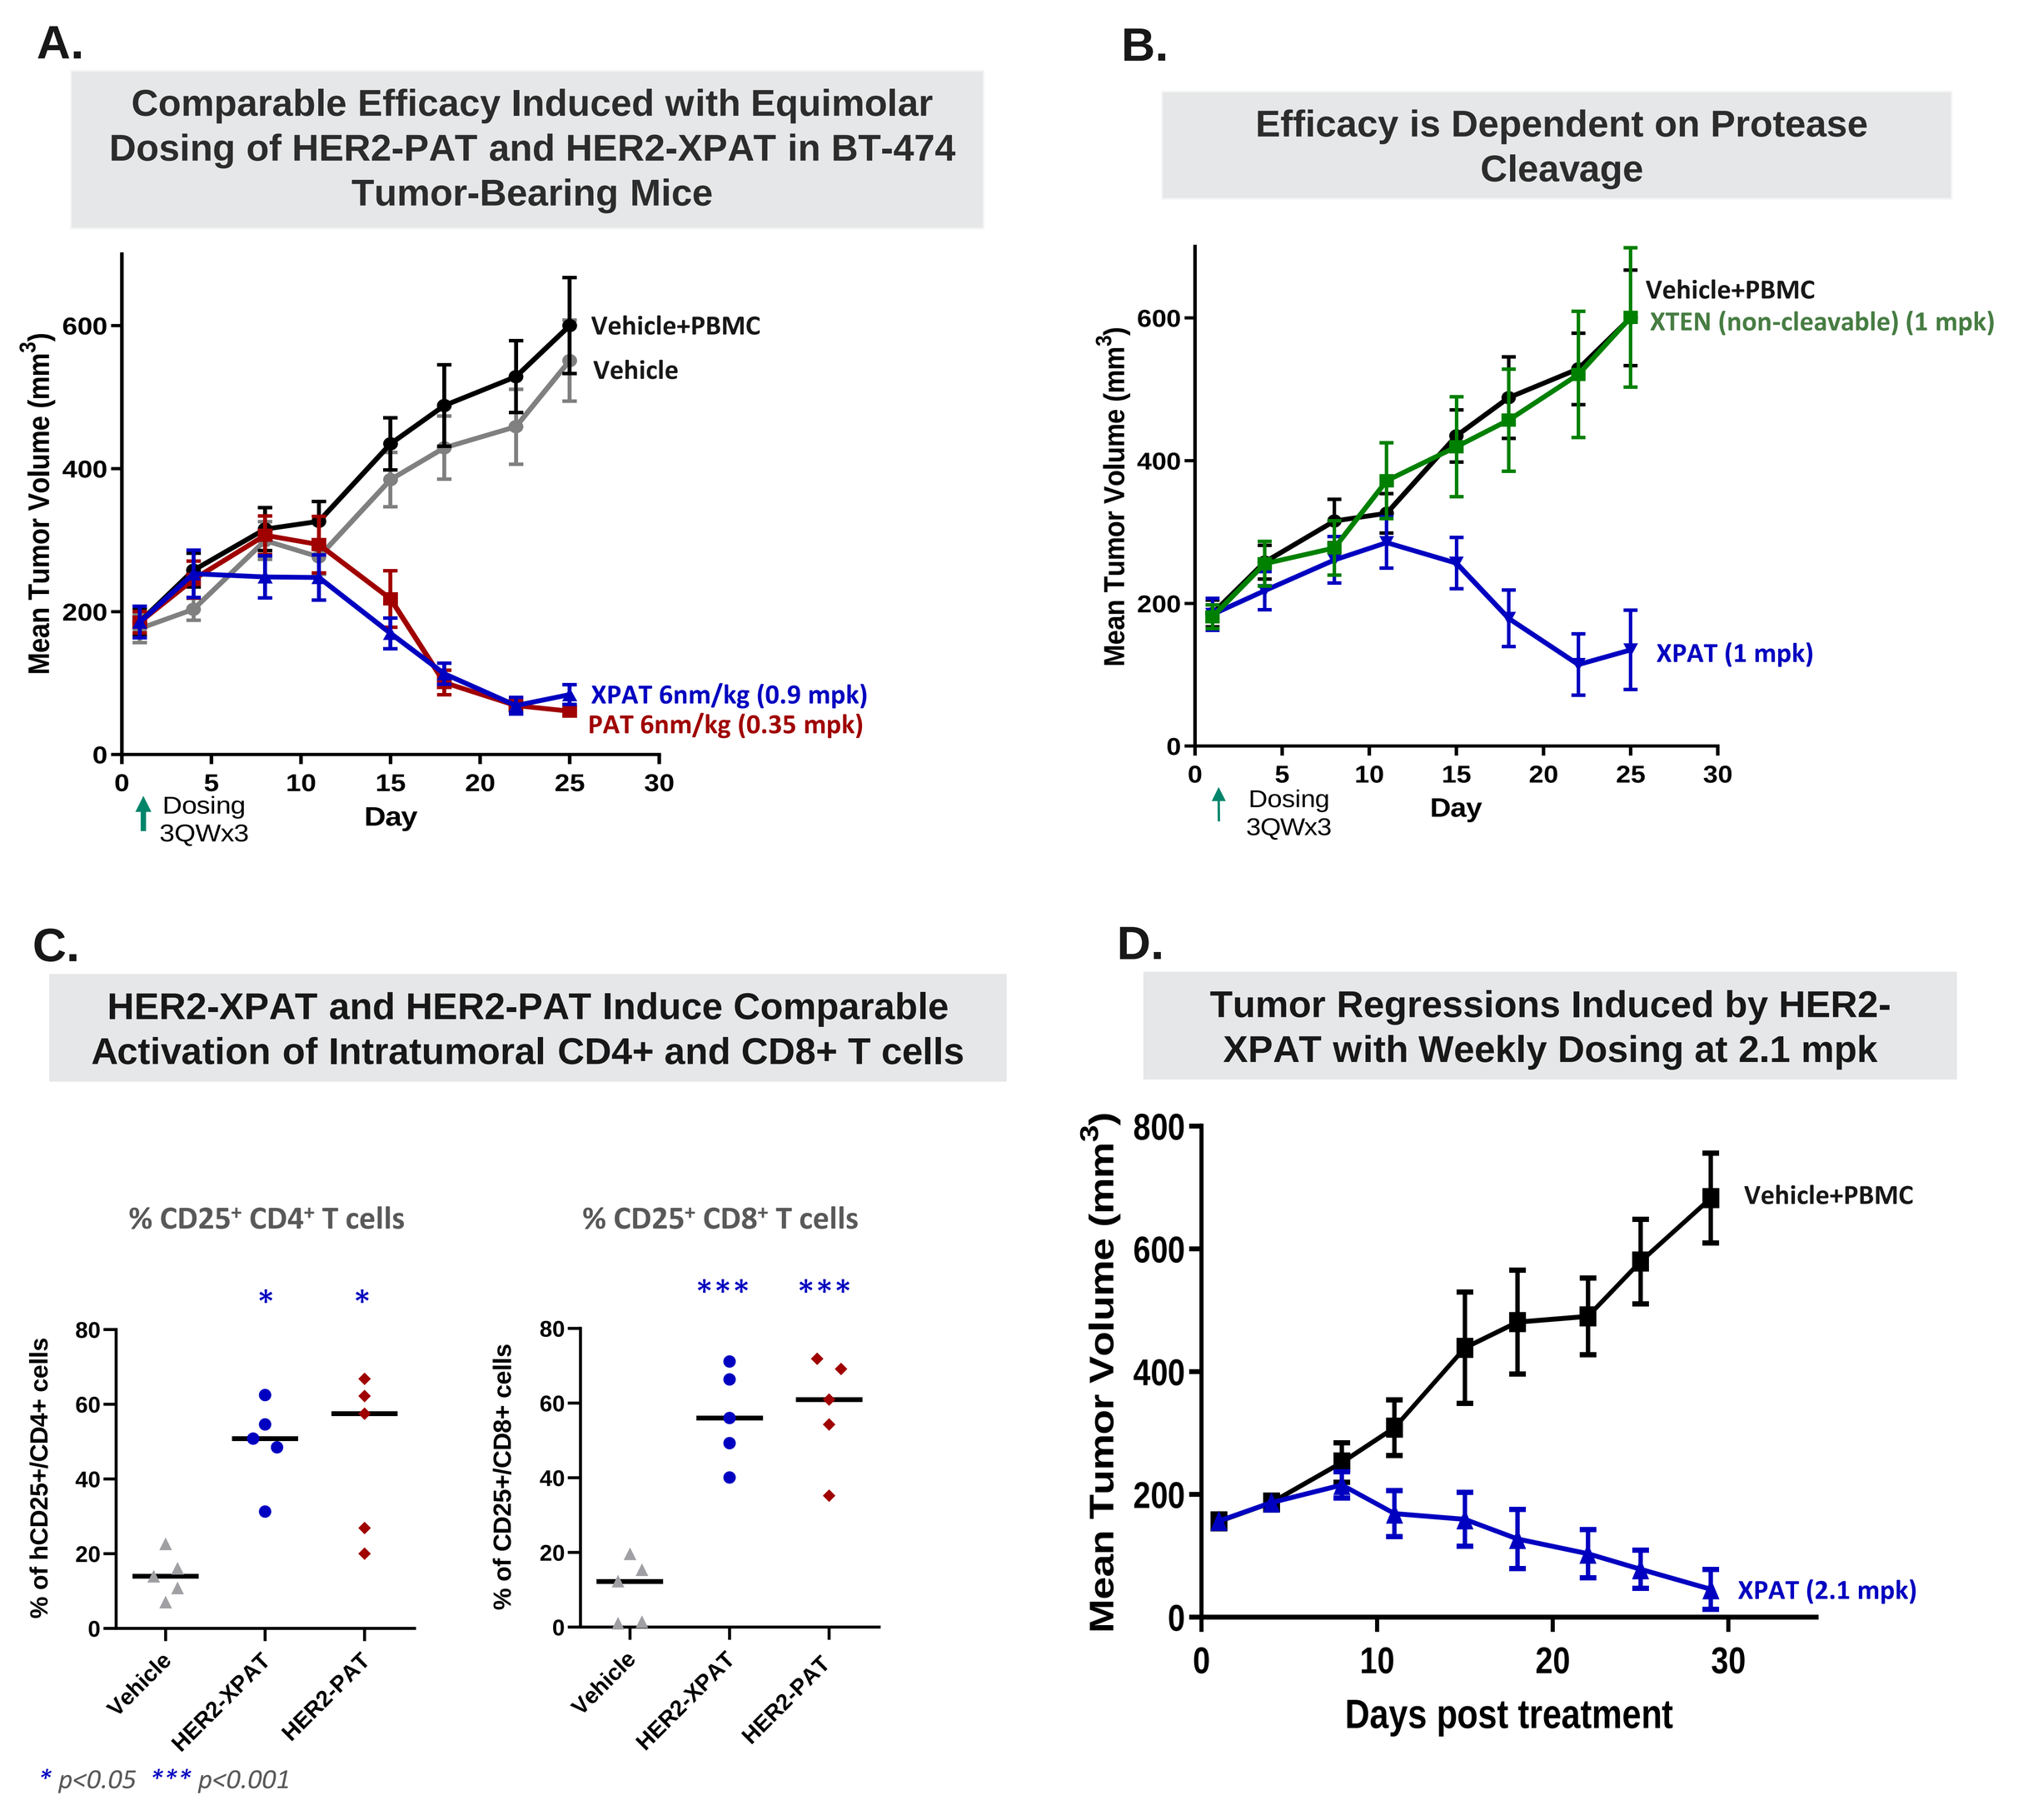
A.
Comparable Efficacy Induced with Equimolar Dosing of HER2-PAT and HER2-XPAT in BT-474 Tumor-Bearing Mice
Vehicle+PBMC
Vehicle
XPAT 6nm/kg (0.9 mpk)
PAT 6nm/kg (0.35 mpk)
Mean Tumor Volume (mm3)
Dosing 3QWx3
Day
B.
Efficacy is Dependent on Protease Cleavage
XTEN (non-cleavable) (1 mpk)
XPAT (1 mpk)
C.
HER2-XPAT and HER2-PAT Induce Comparable Activation of Intratumoral CD4+ and CD8+ T cells
% CD25+ CD4+ T cells
% CD25+ CD8+ T cells
Vehicle
HER2-XPAT
HER2-PAT
* p<0.05 *** p<0.001
D.
Tumor Regressions Induced by HER2-XPAT with Weekly Dosing at 2.1 mpk
XPAT (2.1 mpk)
Days post treatment

A-D) NOG mice were inoculated SC with 2x10⁷ BT-474 tumor cells, engrafted with 1x10⁷ huPBMCs on Day 23 and treated 3 days later with the indicated test articles at equimolar doses TIW for 3 weeks once MTV was 176-192mm³. **B)** C-terminal XTEN lengths on test articles are 50% longer than those used in panels A, C, and D. **C)** Activation status of tumor infiltrating T cells was evaluated by flow cytometry on Day 18 post-TIW dosing of HER2-XPAT (2.1mpk) and HER2-PAT (0.9mpk). **D)** Weekly dosing of HER2-XPAT at 2.1 mpk induced tumor regressions.

Figure 3. XTEN Masks Significantly Expand Safety Margin of HER2-XPAT vs. PAT in Cynomolgus Monkeys

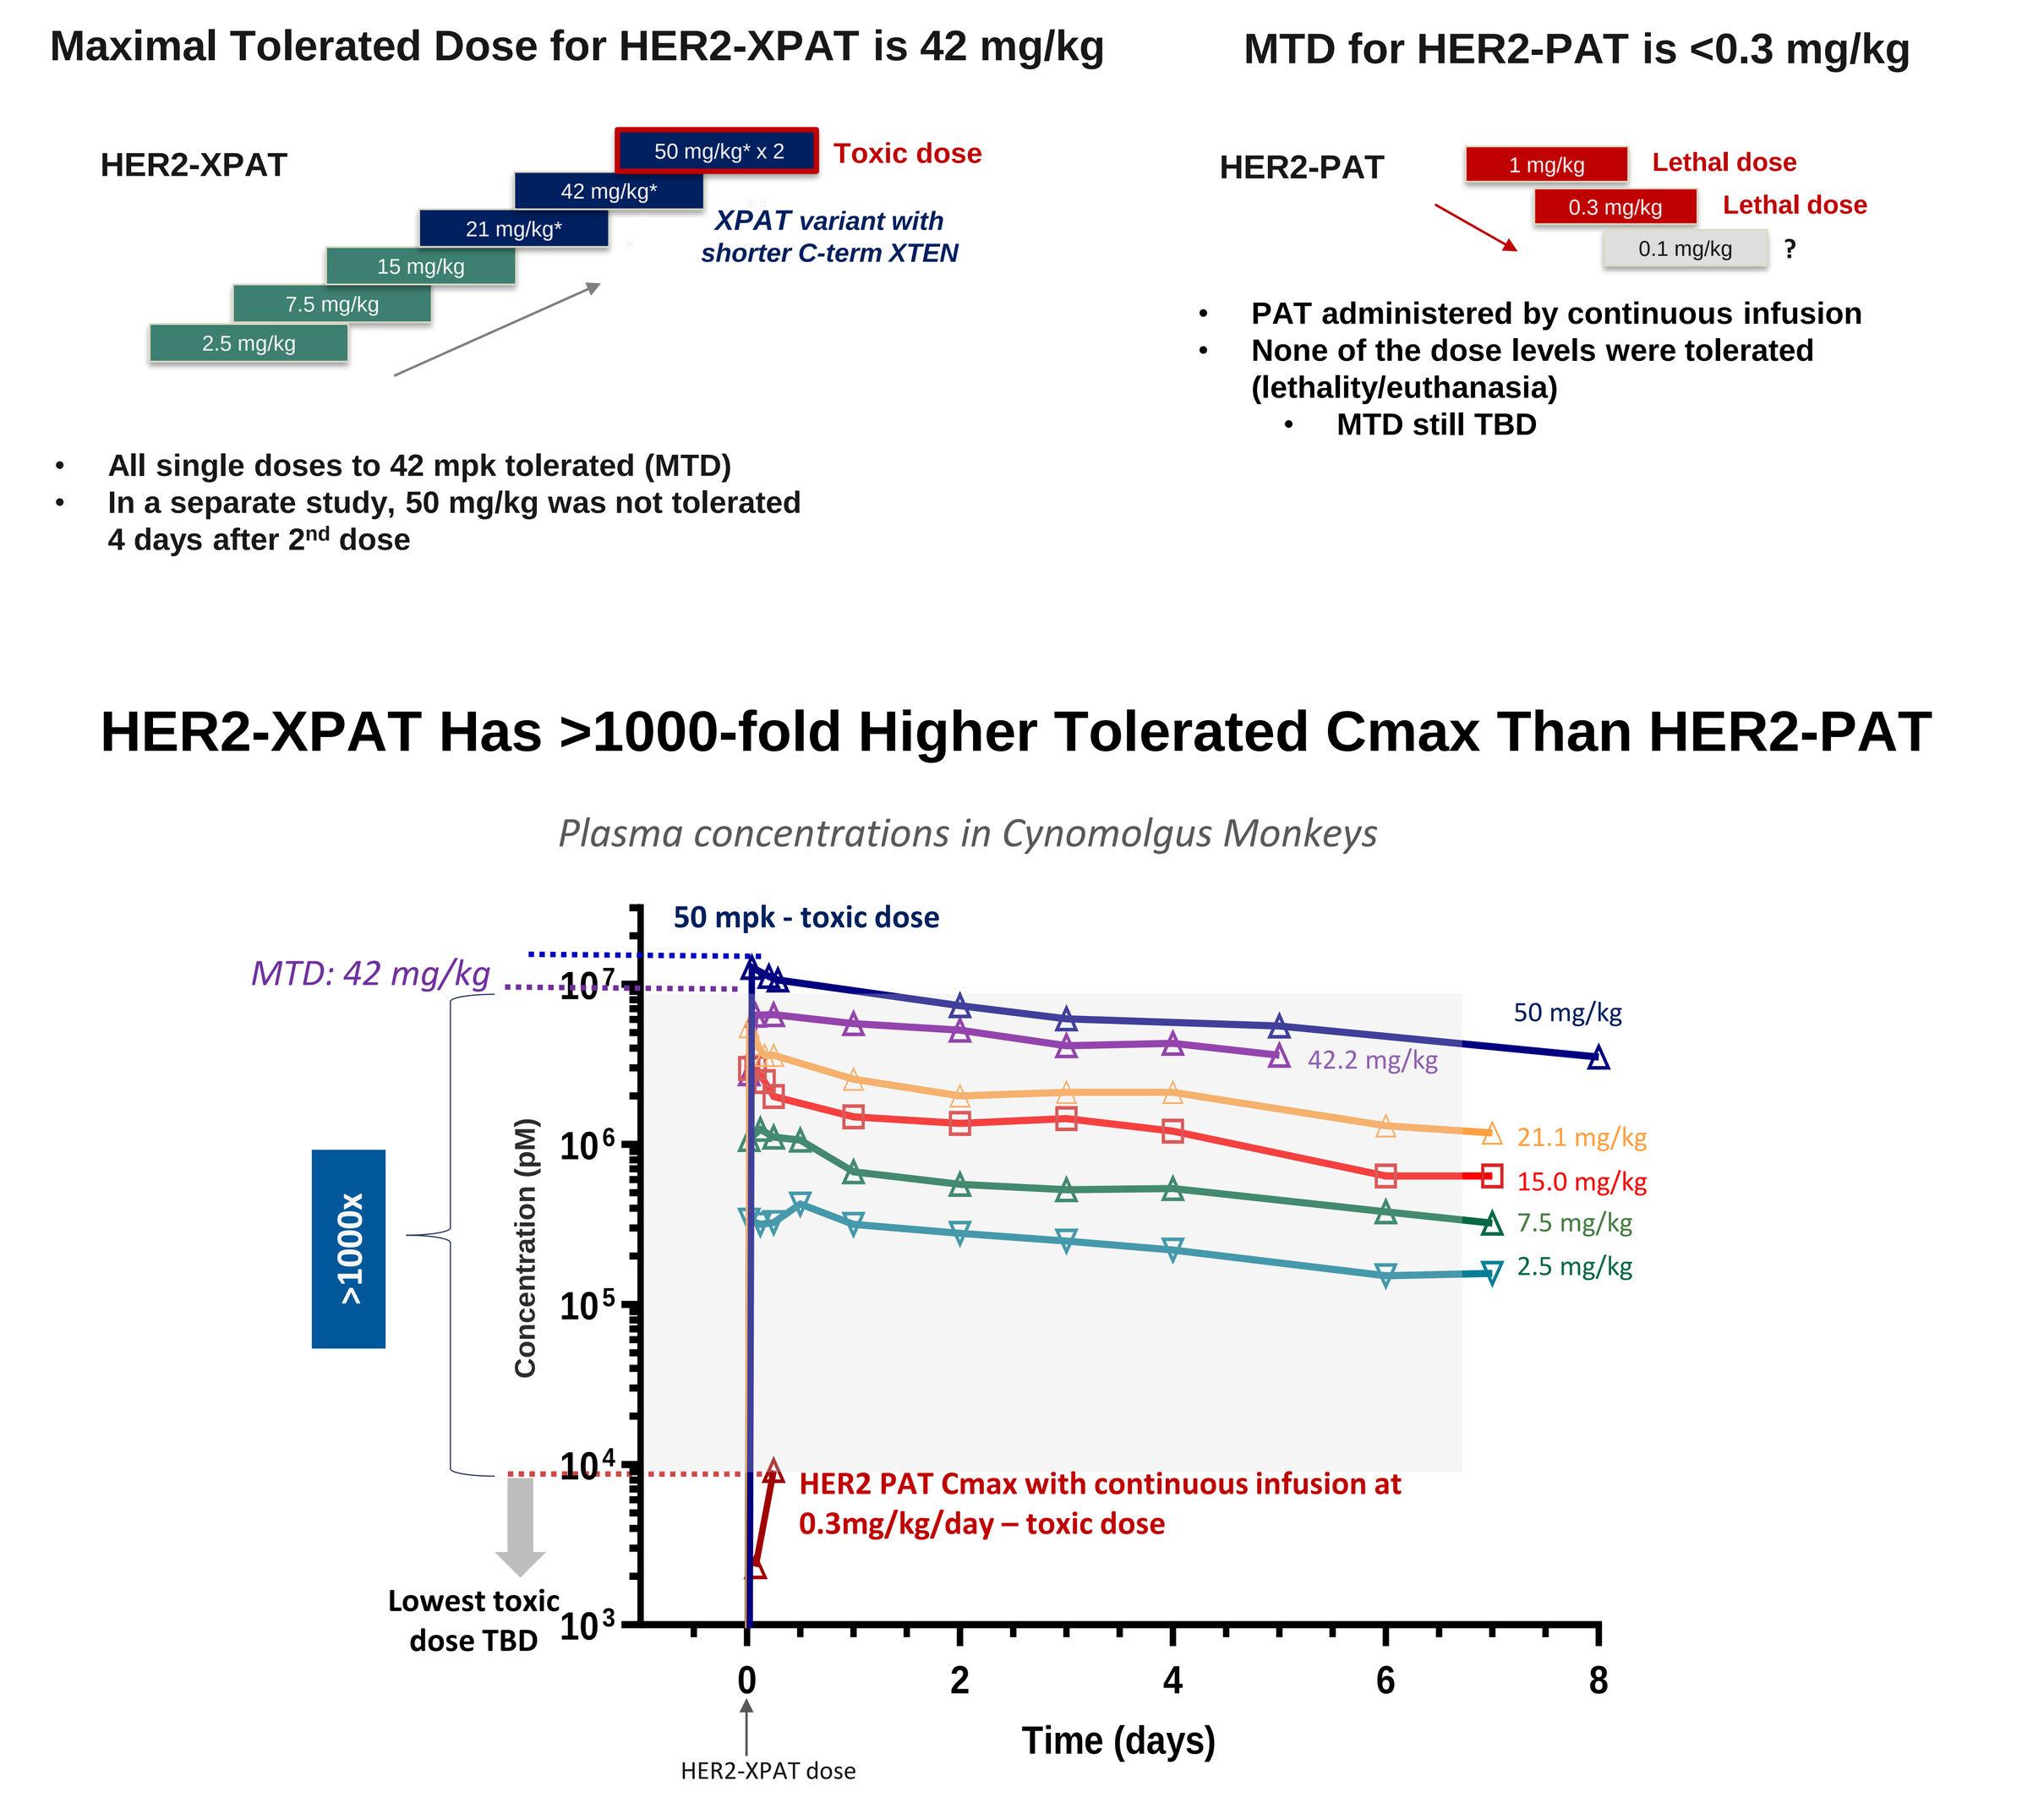
Maximal Tolerated Dose for HER2-XPAT is 42 mg/kg
HER2-XPAT
2.5 mg/kg
7.5 mg/kg
15 mg/kg
21 mg/kg*
42 mg/kg*
50 mg/kg* x 2
Toxic dose
XPAT variant with shorter C-term XTEN
All single doses to 42 mpk tolerated (MTD)
In a separate study, 50 mg/kg was not tolerated 4 days after 2nd dose
MTD for HER2-PAT is <0.3 mg/kg
HER2-PAT
1 mg/kg
0.3 mg/kg
0.1 mg/kg
Lethal dose
?
PAT administered by continuous infusion
None of the dose levels were tolerated (lethality/euthanasia)
MTD still TBD
HER2-XPAT Has >1000-fold Higher Tolerated Cmax Than HER2-PAT
Plasma concentrations in Cynomolgus Monkeys
50 mpk - toxic dose
MTD: 42 mg/kg
>1000x
Concentration (pM)
50 mg/kg
42.2 mg/kg
21.1 mg/kg
15.0 mg/kg
7.5 mg/kg
2.5 mg/kg
HER2 PAT Cmax with continuous infusion at 0.3mg/kg/day – toxic dose
Lowest toxic dose TBD
HER2-XPAT dose
Time (days)

HER2 XPAT was administered IV, single dose/animal (doses 2.5-42mpk) and weekly x2 at 50mpk. *At doses 21mpk and above, a variant of HER2-XPAT with a shorter C-terminal XTEN mask was used. HER2-PAT was administered by continuous infusion due to its short half-life. Plasma concentrations of HER2-XPAT were measured by ELISA using recombinant HER2 capture and an antibody directed against the XTEN mask for detection. For measurement of HER2-PAT concentrations, a peptide encoding the N-terminus of CD3 epsilon was used for detection.

Figure 4. HER2-XPAT Induces T Cell Margination at Doses >2.5 mpk

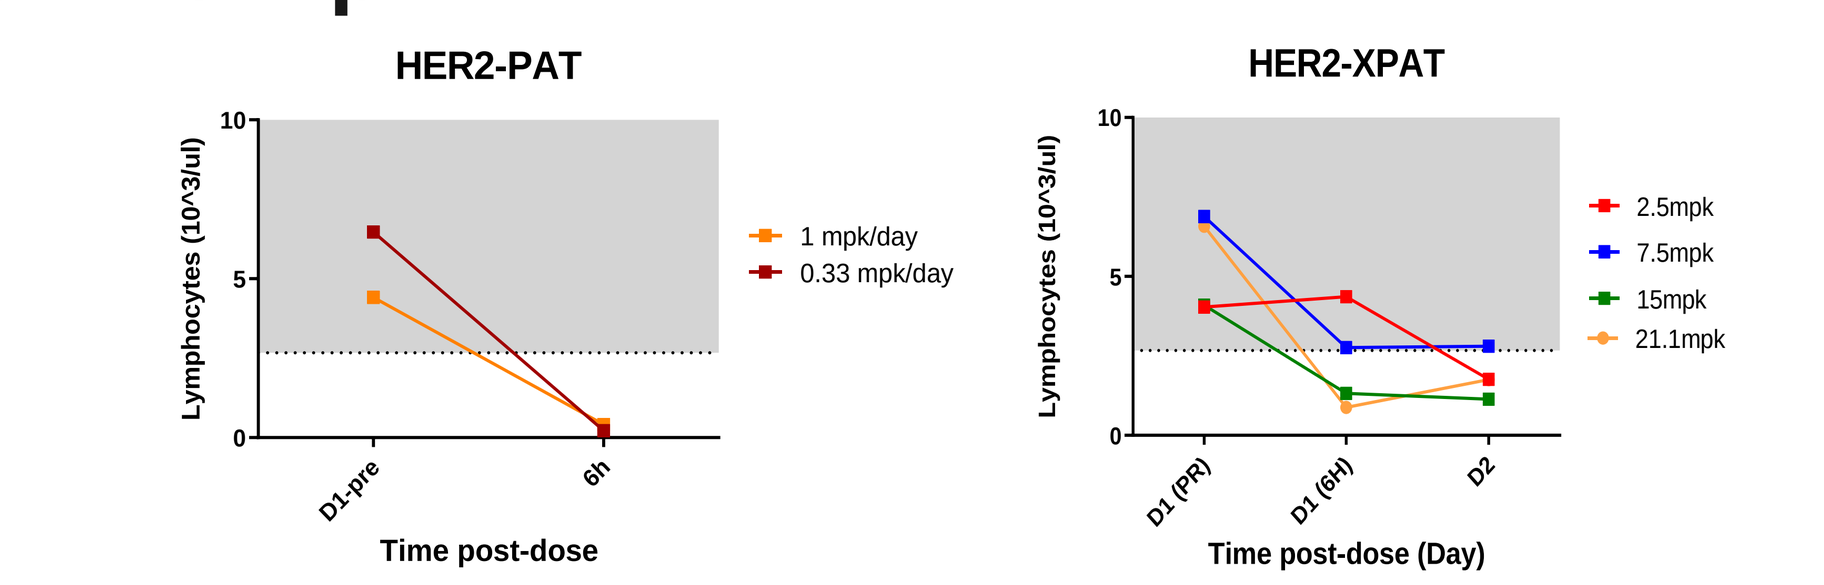
HER2-PAT
HER2-XPAT
Lymphocytes (10^3/ul)
1 mpk/day
0.33 mpk/day
2.5mpk
7.5mpk
15mpk
21.1mpk
Time post-dose
Time post-dose (Day)

Blood Lymphocyte counts decline sharply at 6 or 24 hours post-dose, consistent with T cell margination

Figure 5. HER2-XPAT Fails to Activate Peripheral T Cells or Induce Cytokine Release Syndrome in NHPs, Even at 50 mg/kg Doses; HER2-PAT Induces CRS at 0.3 mg/kg

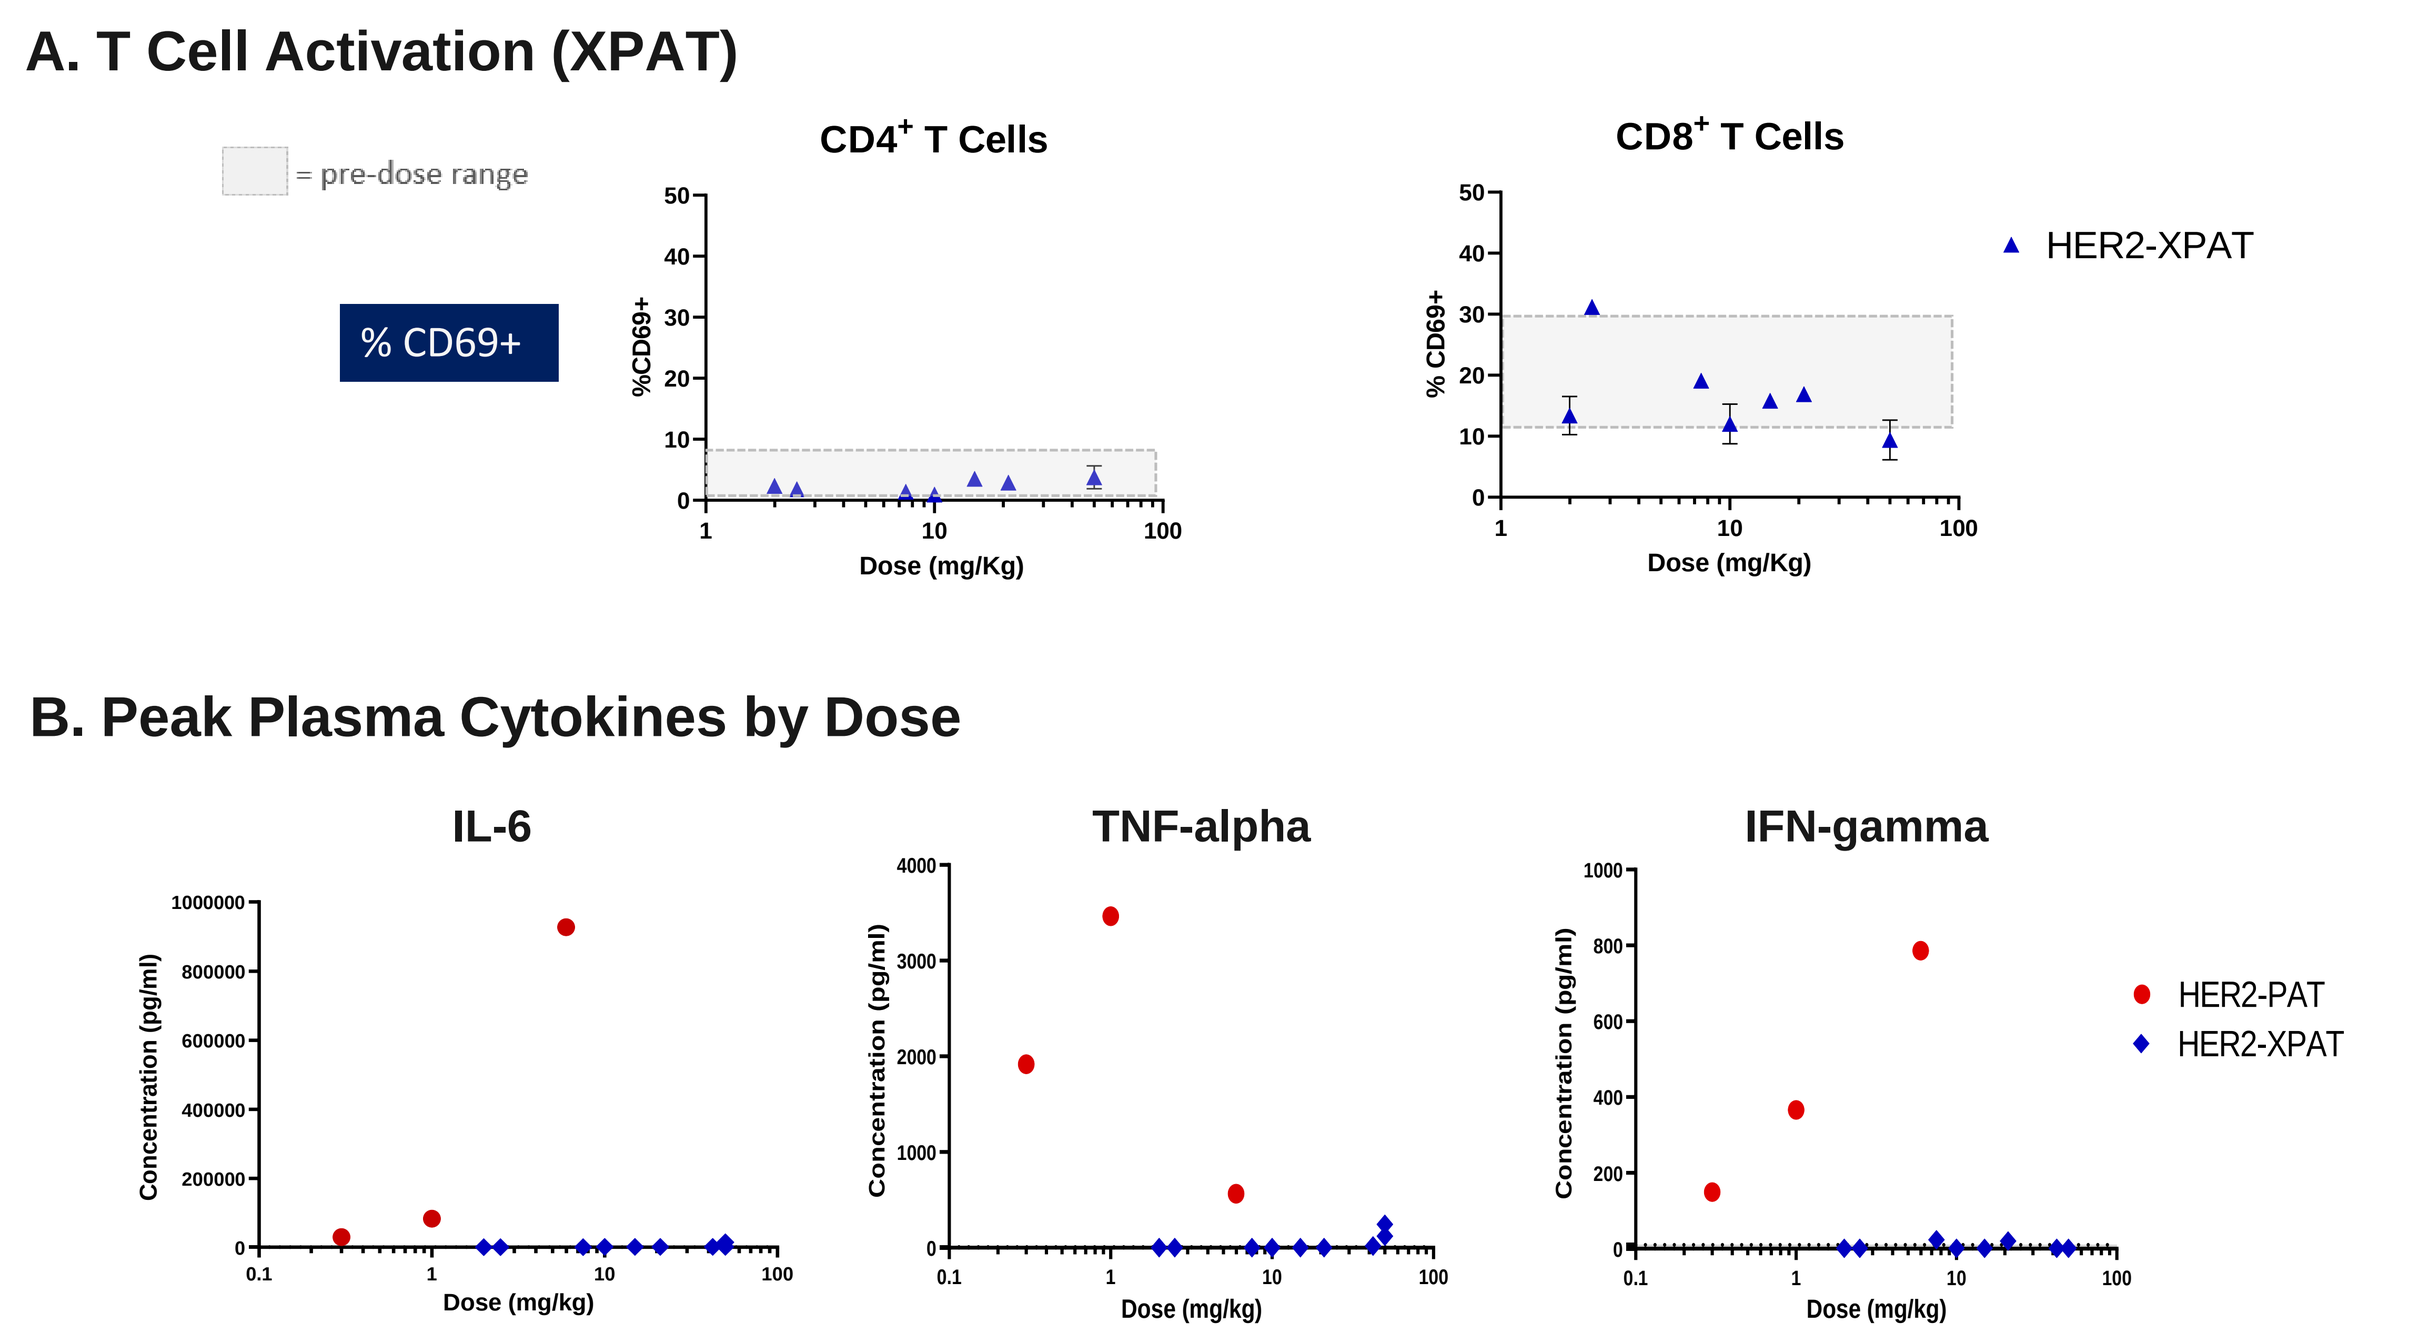
A. T Cell Activation (XPAT)
= pre-dose range
% CD69+
CD4+ T Cells
CD8+ T Cells
HER2-XPAT
Dose (mg/Kg)
B. Peak Plasma Cytokines by Dose
IL-6
TNF-alpha
IFN-gamma
Concentration (pg/ml)
Dose (mg/kg)
HER2-PAT
HER2-XPAT

A) Peripheral T cell activation (%CD69⁺) was evaluated by flow cytometry 24 hours post-HER2-XPAT treatment. **B)** Cytokine analysis was performed with a Luminex® suspension array system on plasma samples. Data presented are maximal values measured between 6-24 hours at each evaluated dose.

Figure 6. XPATs Can Significantly Expand the Therapeutic Index of TCEs Directed Against Broadly Expressed Targets: EGFR

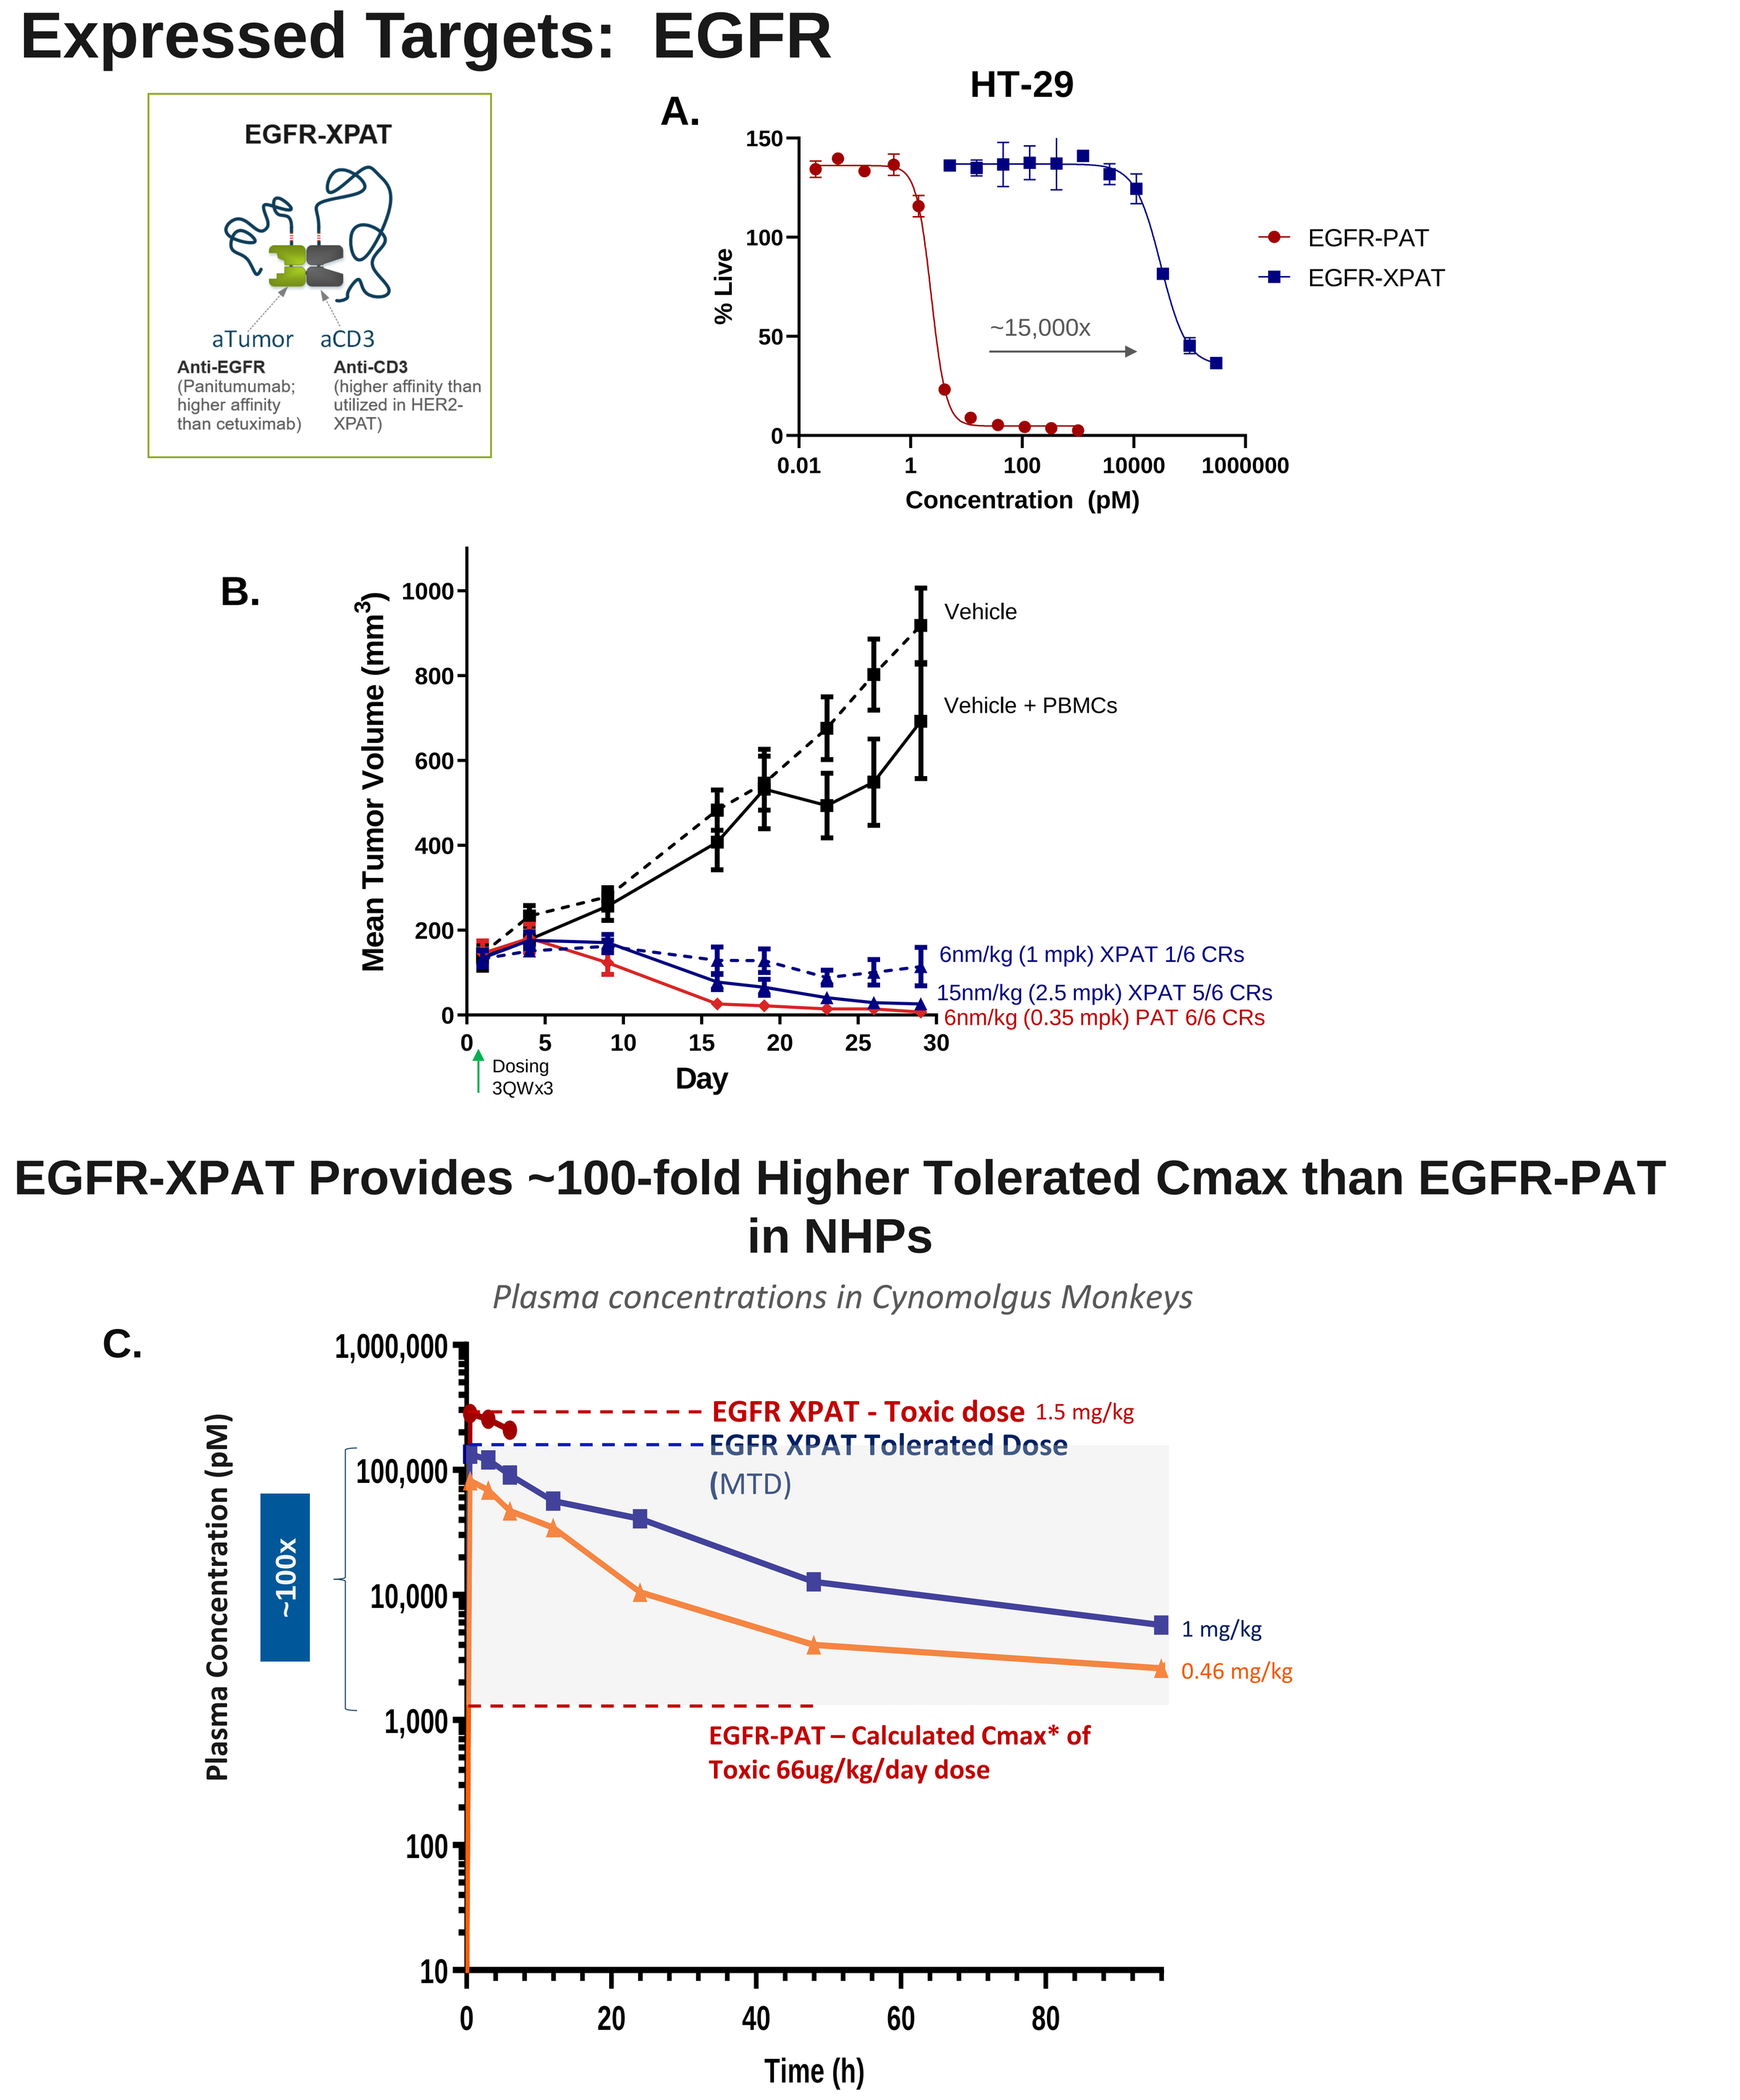
EGFR-XPAT
aTumor
aCD3
Anti-EGFR (Panitumumab; higher affinity than cetuximab)
Anti-CD3 (higher affinity than utilized in HER2-XPAT)
A.
HT-29
% Live
~15,000x
EGFR-PAT
EGFR-XPAT
Concentration (pM)
B.
Vehicle
Vehicle + PBMCs
6nm/kg (1 mpk) XPAT 1/6 CRs
15nm/kg (2.5 mpk) XPAT 5/6 CRs
6nm/kg (0.35 mpk) PAT 6/6 CRs
Mean Tumor Volume (mm3)
Dosing 3QWx3
Day
EGFR-XPAT Provides ~100-fold Higher Tolerated Cmax than EGFR-PAT in NHPs
Plasma concentrations in Cynomolgus Monkeys
C.
EGFR XPAT - Toxic dose 1.5 mg/kg
EGFR XPAT Tolerated Dose (MTD)
~100x
1 mg/kg
0.46 mg/kg
EGFR-PAT – Calculated Cmax* of Toxic 66ug/kg/day dose
Plasma Concentration (pM)
Time (h)

A) Cytotoxicity was quantified using Cell Titer-Glo cell viability assay following a 48 hour incubation of huPBMCs and HT-29 cells at a 10:1 ratio. **B)** NOG mice were implanted SC with 1x10⁷ HT-29 tumor cells, engrafted with 1x10⁷ huPBMCs on Day15 and treated with test articles at the indicated doses TIW for 3 weeks once tumors reached ~150mm3. **C)** In NHP, EGFR-XPAT was administered IV at the indicated doses; EGFR-PAT was administered by continuous infusion at 66ug/kg/day following a 1 hour bolus dose of 8ug/kg. Plasma concentrations of EGFR-XPAT were measured by ELISA using recombinant EGFR as capture and an a-XTEN antibody for detection. *The Cmax value for EGFR PAT is calculated, awaiting analysis of PK samples for confirmation.

SUMMARY/CONCLUSIONS

- *In vitro*, proteolytically-unmasked HER2- and EGFR-XPATs demonstrate potent cytotoxicity against tumor lines with EC50s in the single-digit pM range
 - XTEN masking reduces target-directed T cell cytotoxicity and T cell activation by up to 15,000-fold
- In established xenograft models, HER2 and EGFR XPATs induced protease-dependent tumor regressions with efficacious doses within an order of magnitude of the unmasked (active) T cell engager
- In cynomolgus monkeys, masked XPATs demonstrated significantly reduced CRS and increased tolerated exposures relative to unmasked PAT, suggesting favorable therapeutic index even for targets as broadly expressed as EGFR
 - HER2- and EGFR-XPATs had maximum tolerated exposures that were >1000-fold and ~100-fold higher than those of their respective active forms (PAT)
- XPATs represent a novel strategy to improve the toxicity profile of T cell engagers while maintaining their potency against solid tumors, thus enabling a significant increase in the therapeutic index and expansion of target landscape for this potent modality