HER2-XPAT, A Novel Protease-Activatable Prodrug T Cell Engager (TCE), Engineered to Address On-Target, Off Tumor Toxicity and Provide Large Predicted Safety Margins in Non-Human Primates

Fiore Cattaruzza, Ayesha Nazeer, Zachary Lange, Caitlin Koski, Mikhail Hammond, Trang Dao-Pick, Angela Henkensiefken, Daniel Hostetter, Mika K. Derynck, Volker Schellenberger and Bryan A. Irving

Amunix Pharmaceuticals, Inc. Mountain View, CA

INTRODUCTION

Bispecific T Cell Engagers (TCEs) are effective at inducing remissions in hematologic cancers, but their use in solid tumors has been challenging due to their extreme potency and on target, off-tumor toxicities in healthy tissue. To address this challenge, Amunix has developed conditionally active TCEs, XPATs or **X**TENylated **P**rotease-**A**ctivated bispecific **T** Cell Engagers targeting HER2 and EGFR that exploit the dysregulated protease activity present in tumors vs. healthy tissues, enabling expansion of the therapeutic index. The XPAT core consists of 2 single chain antibody fragments (scFvs) targeting CD3 and the tumor target. Two unstructured polypeptide masks (XTEN) are attached to the core that sterically reduce target engagement and extend protein half-life. Protease cleavage sites at the base of the XTEN masks enable proteolytic activation of XPAT in the tumor microenvironment, unleashing a small, highly potent TCE. In healthy tissues, where protease activity is tightly regulated, XPATs should remain predominantly inactive as intact prodrugs. In addition to localized activation, the short half-life of the unmasked PAT form should further widen the therapeutic index while providing the potency of T-cell immunity to improve the eradication of solid tumors.

XPAT PLATFORM

XPATs Are XTENylated Protease-Activated T Cell Engagers

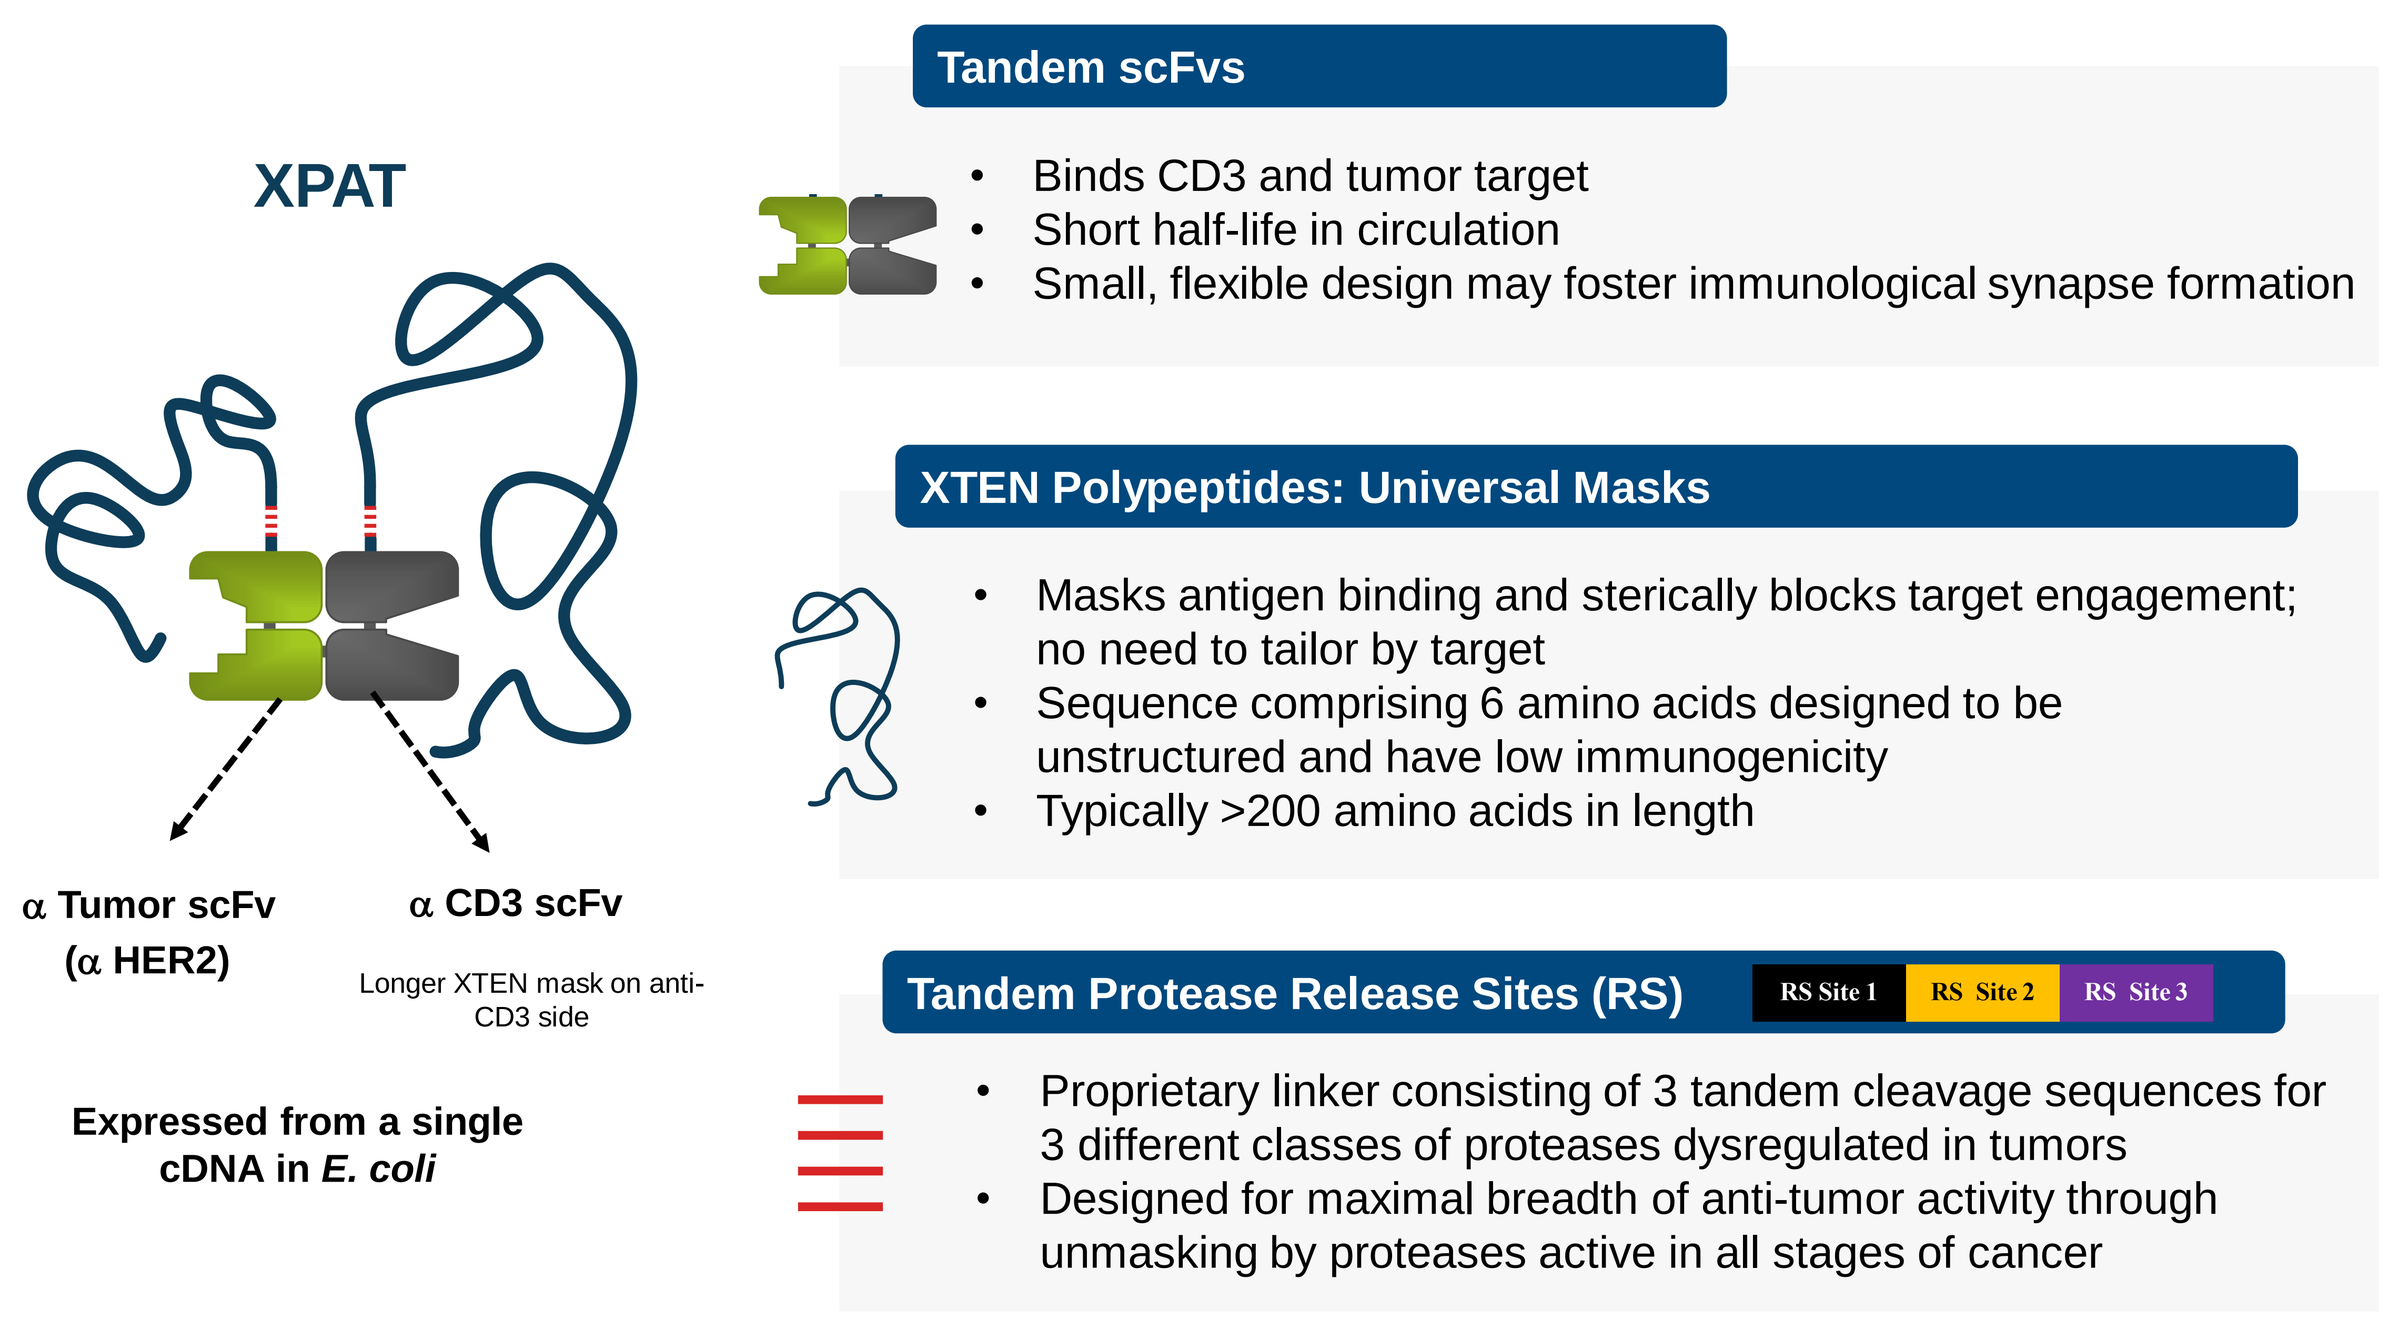
XPAT
α Tumor scFv (α HER2)
α CD3 scFv
Longer XTEN mask on anti-CD3 side
Expressed from a single cDNA in *E. coli*
Tandem scFvs
• Binds CD3 and tumor target
• Short half-life in circulation
• Small, flexible design may foster immunological synapse formation
XTEN Polypeptides: Universal Masks
• Masks antigen binding and sterically blocks target engagement; no need to tailor by target
• Sequence comprising 6 amino acids designed to be unstructured and have low immunogenicity
• Typically >200 amino acids in length
Tandem Protease Release Sites (RS)
RS Site 1
RS Site 2
RS Site 3
• Proprietary linker consisting of 3 tandem cleavage sequences for 3 different classes of proteases dysregulated in tumors
• Designed for maximal breadth of anti-tumor activity through unmasking by proteases active in all stages of cancer

XPATs Enable Localized Tumor Killing, Limiting Toxicity Against Healthy Tissue Expressing the Target Antigen

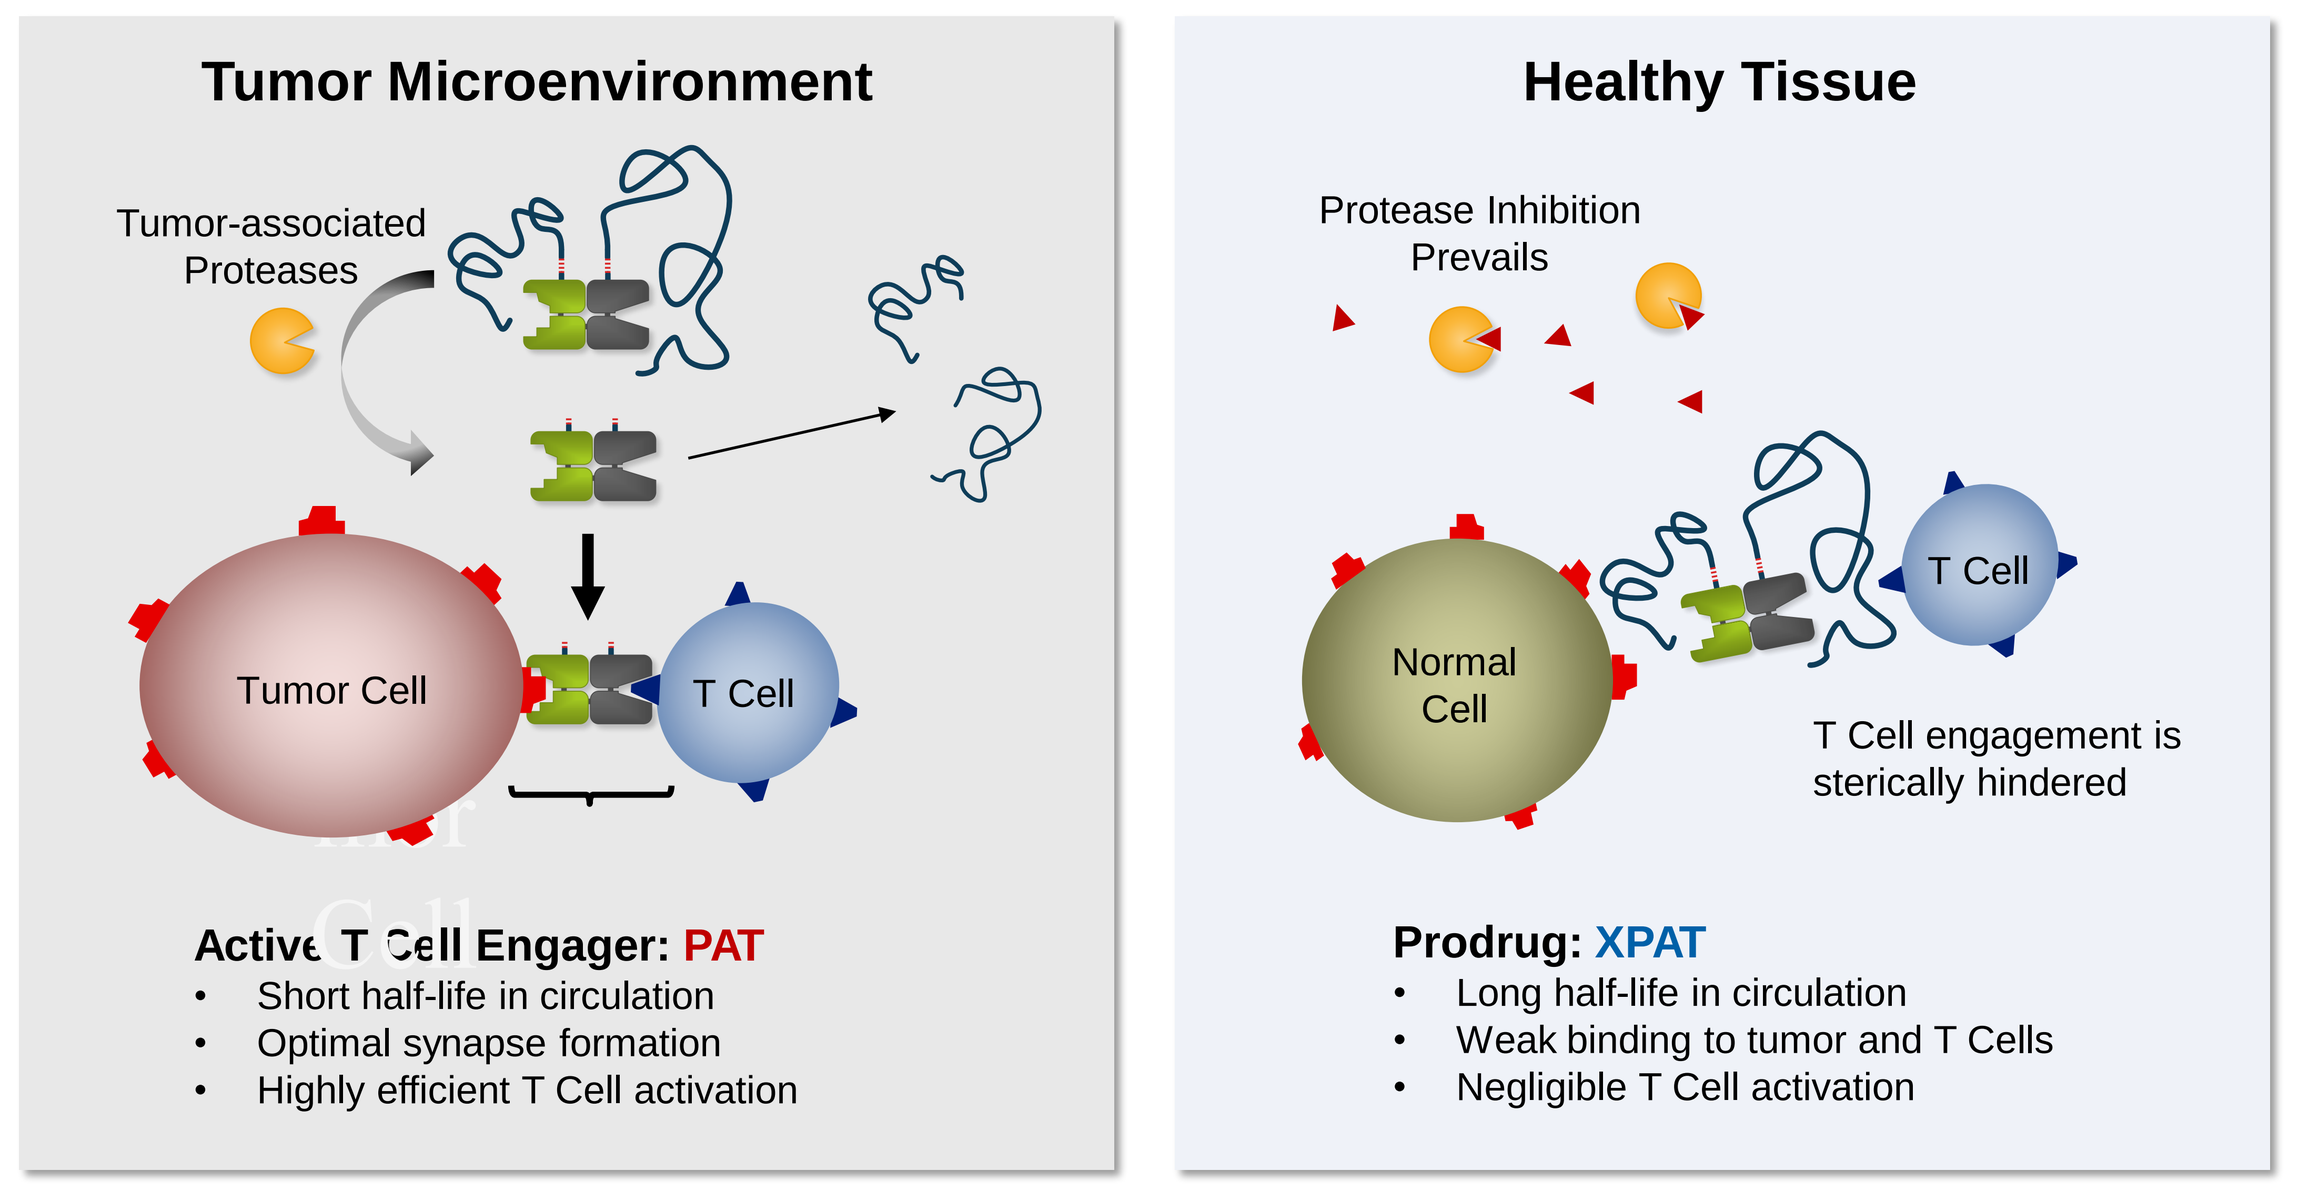
Tumor Microenvironment
Tumor-associated Proteases
Tumor Cell
T Cell
Active T Cell Engager: PAT
• Short half-life in circulation
• Optimal synapse formation
• Highly efficient T Cell activation
Healthy Tissue
Protease Inhibition Prevails
Normal Cell
T Cell
T Cell engagement is sterically hindered
Prodrug: XPAT
• Long half-life in circulation
• Weak binding to tumor and T Cells
• Negligible T Cell activation

RESULTS

Figure 1. XTEN Polypeptide Masks on HER2-XPAT Significantly Reduce T Cell–Mediated Cytotoxicity and T Cell Activation *in vitro*

A. Tumor-directed Cytotoxicity

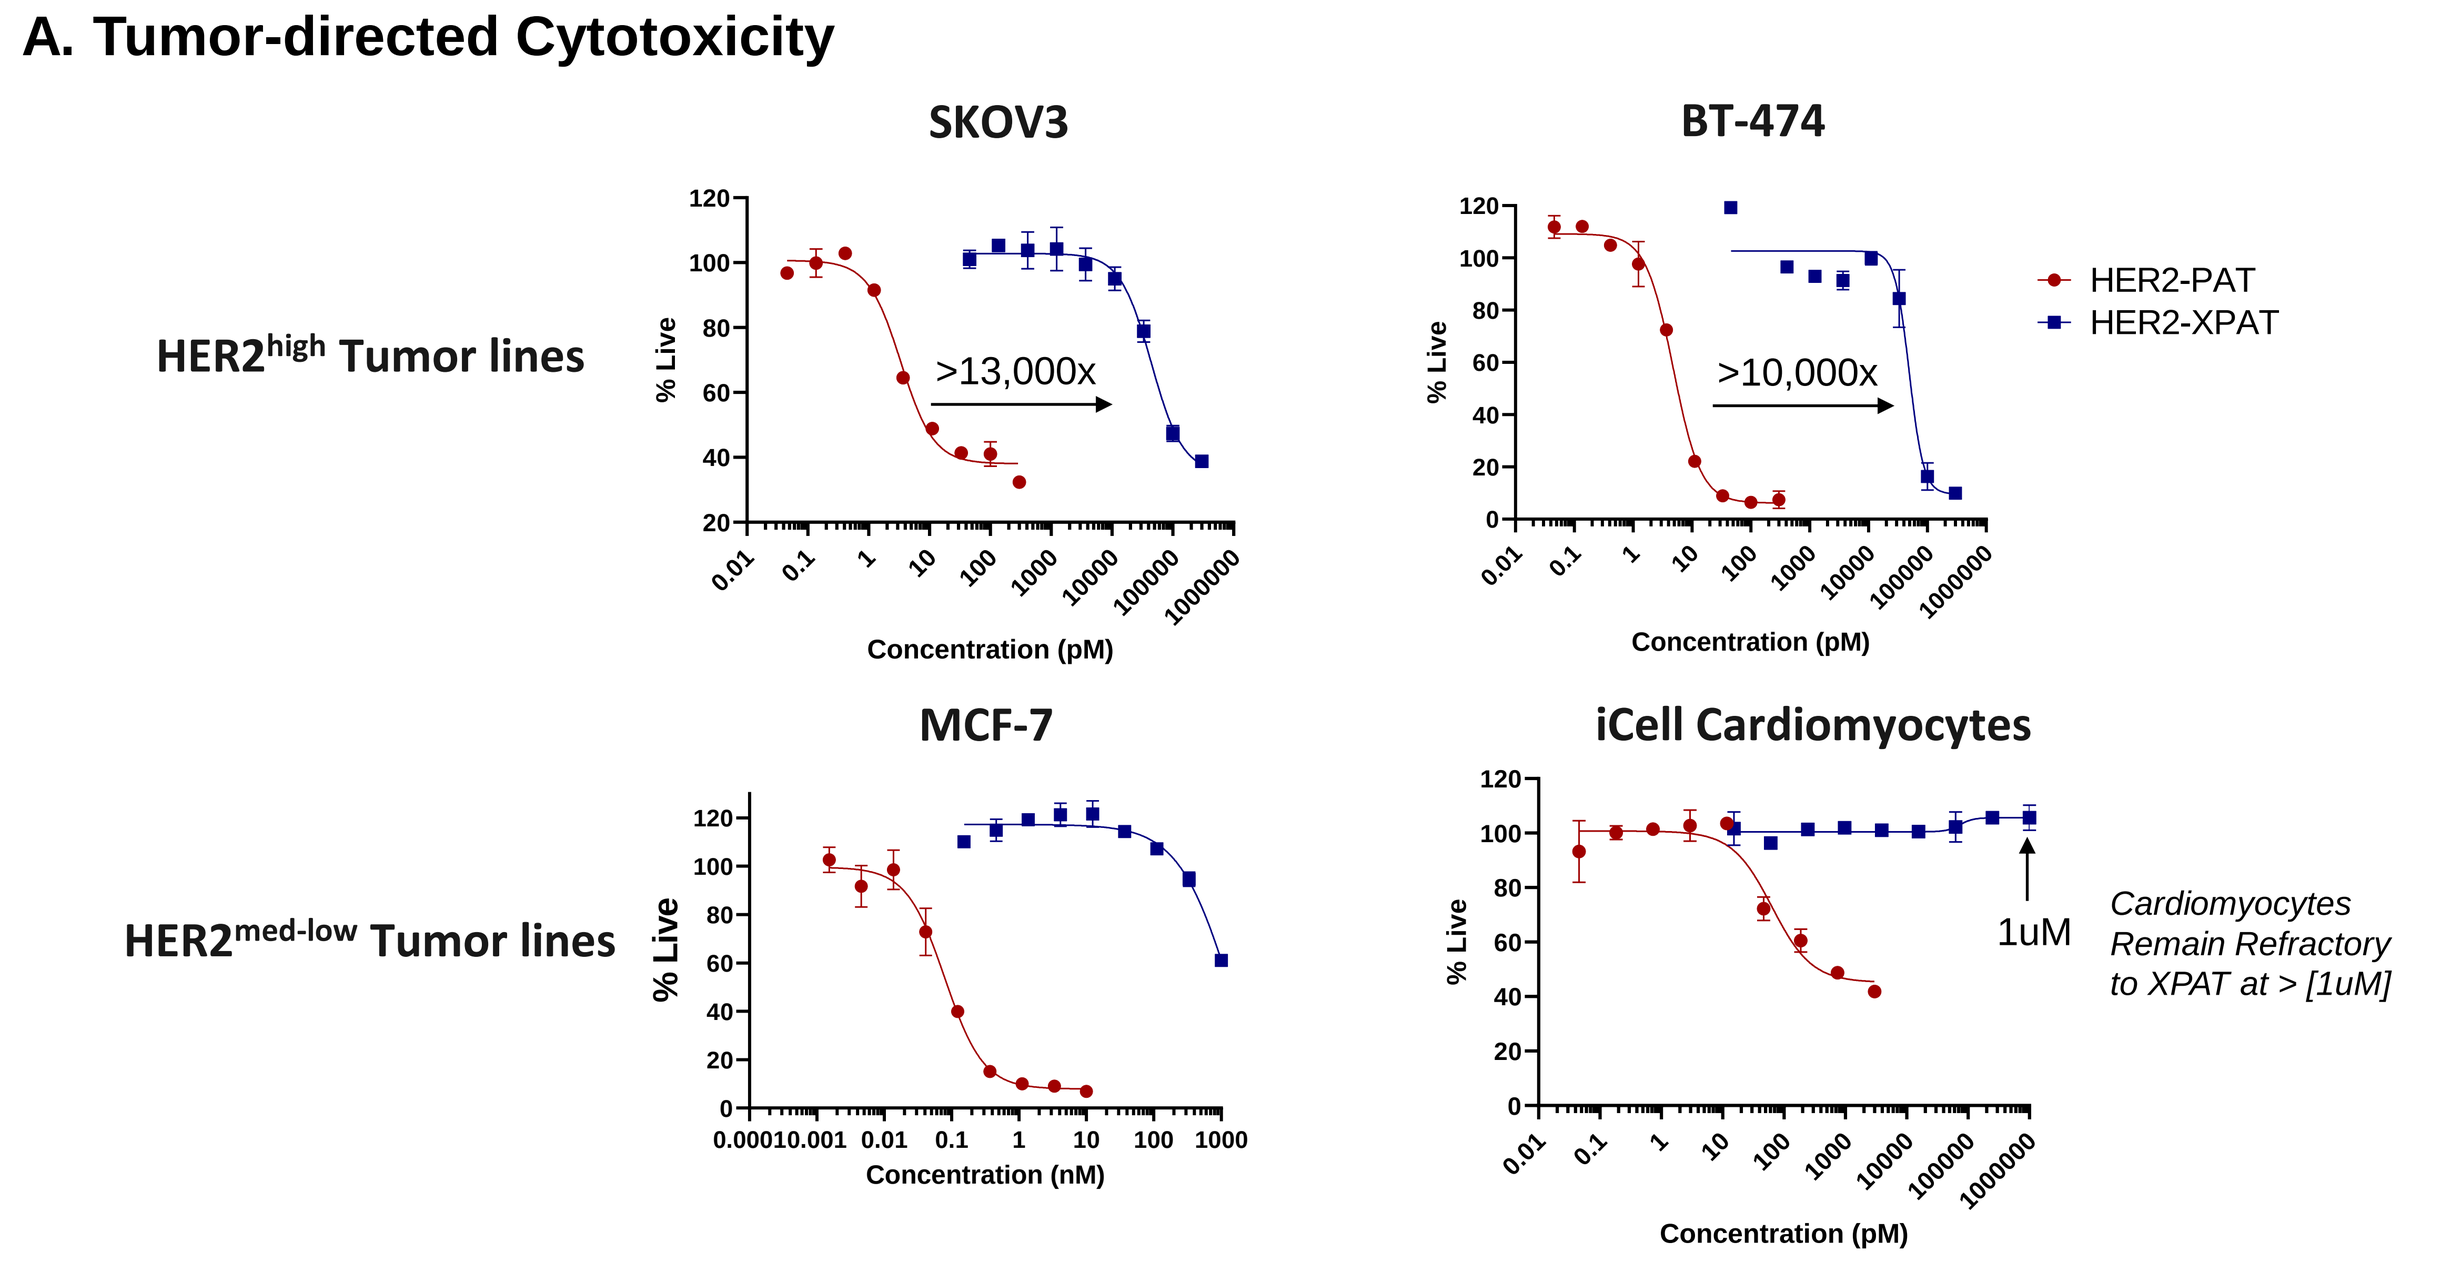
SKOV3
BT-474
HER2high Tumor lines
>13,000x
>10,000x
MCF-7
iCell Cardiomyocytes
HER2med-low Tumor lines
1uM
Cardiomyocytes Remain Refractory to XPAT at > [1uM]
% Live
Concentration (pM)
Concentration (nM)
HER2-PAT
HER2-XPAT

B. Target-dependent T Cell Activation

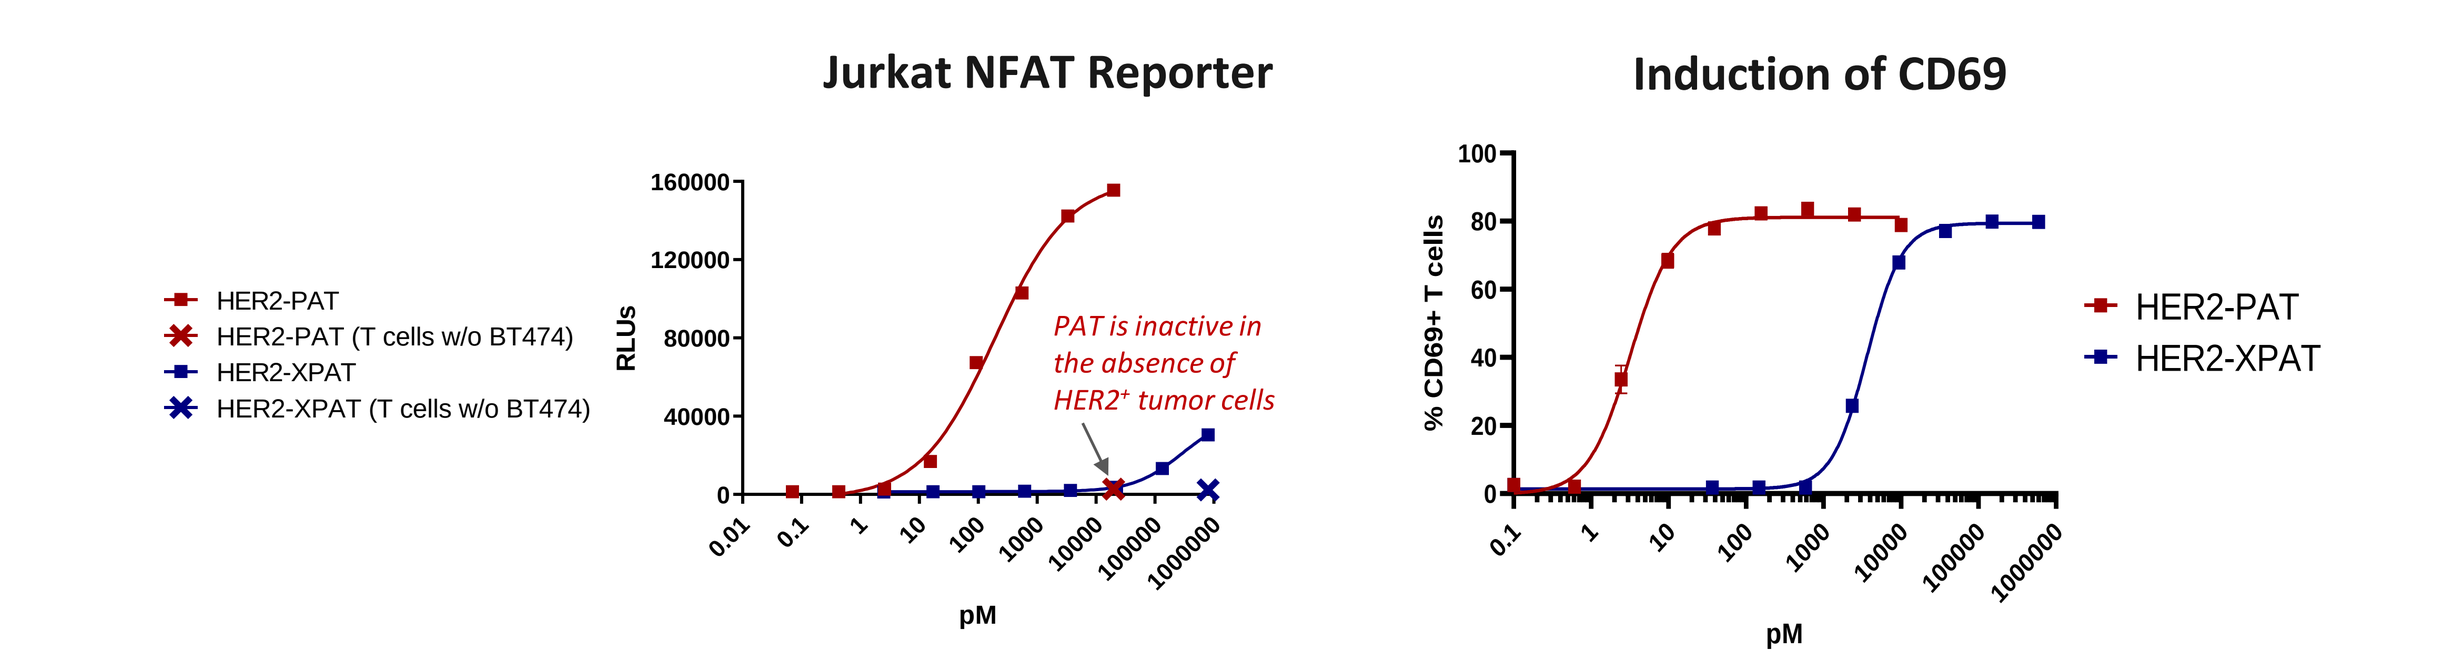
Jurkat NFAT Reporter
Induction of CD69
HER2-PAT
HER2-PAT (T cells w/o BT474)
HER2-XPAT
HER2-XPAT (T cells w/o BT474)
PAT is inactive in the absence of HER2+ tumor cells
RLUs
% CD69+ T cells
pM

A) Cytotoxicity was quantified using Cell Titre-Glo Luminescent Cell Viability Assay following a 48 hour incubation of huPBMCs and the indicated tumor cell lines or human iCell Cardiomyocytes at a 1:1 Effector:Target ratio. Co-cultures were treated with HER2-XPAT or HER2-PAT at the concentrations shown. **B)** Jurkat reporter T cells were incubated with or without BT-474 cells at a 5:1 E:T ratio for 6 hours and NFAT-induced Luciferase activity quantified in response to the test articles. Surface CD69 expression was evaluated on T cells by flow cytometry following a 72 hour co-incubation of PBMCs and SKOV3 cells at a 5:1 E:T ratio with test articles at the indicated concentrations.

Figure *2.* HER2-XPAT Induces Robust Tumor Regressions in Mice That are Dependent on the Protease Release Site

A. Comparable efficacy induced with equimolar dosing of HER2-PAT and HER2-XPAT in BT-474 tumor-bearing mice

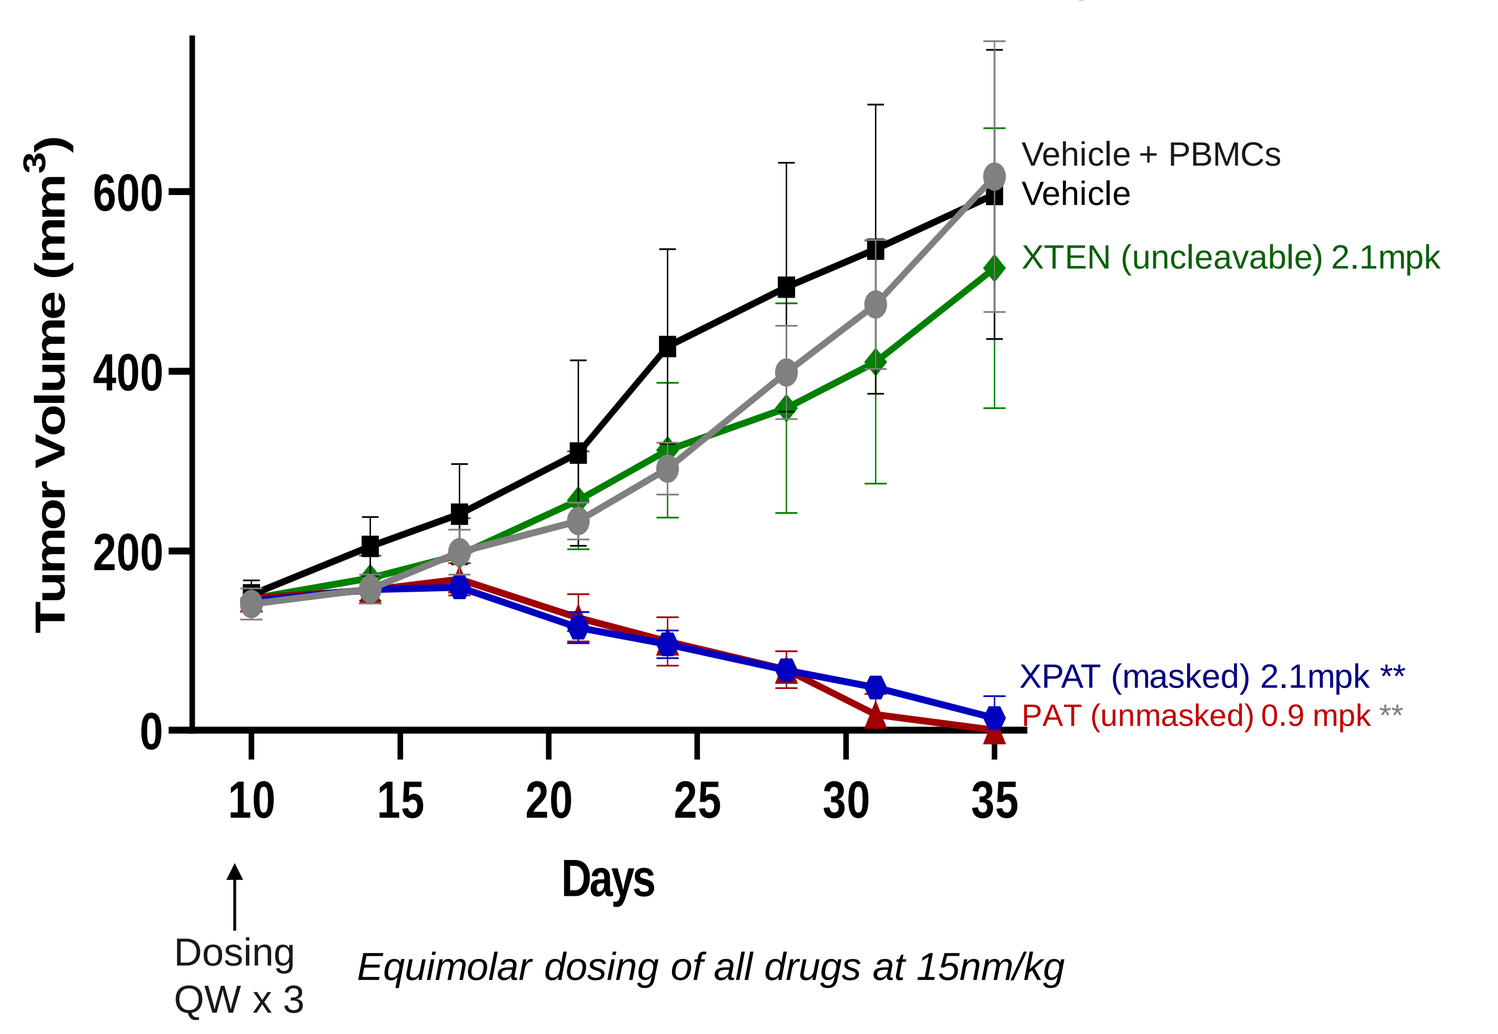
Tumor Volume (mm3)
Days
Vehicle + PBMCs
Vehicle
XTEN (uncleavable) 2.1mpk
XPAT (masked) 2.1mpk **
PAT (unmasked) 0.9 mpk **
Dosing QW x 3
Equimolar dosing of all drugs at 15nm/kg

B. HER2-XPAT is effective against large tumors with a single dose

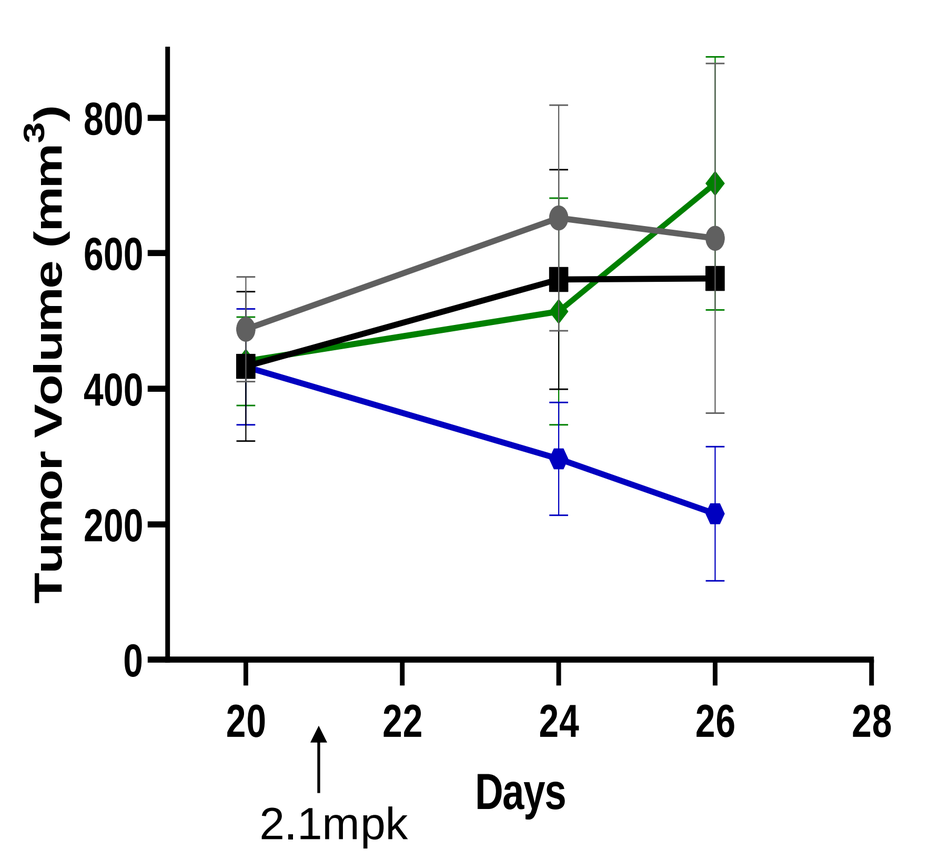
Tumor Volume (mm3)
Days
2.1mpk

C. HER2-XPAT and HER2-PAT induce comparable activation of intratumoral CD4+ and CD8+ T cells

Activation not observed in the periphery in the absence of huHER2

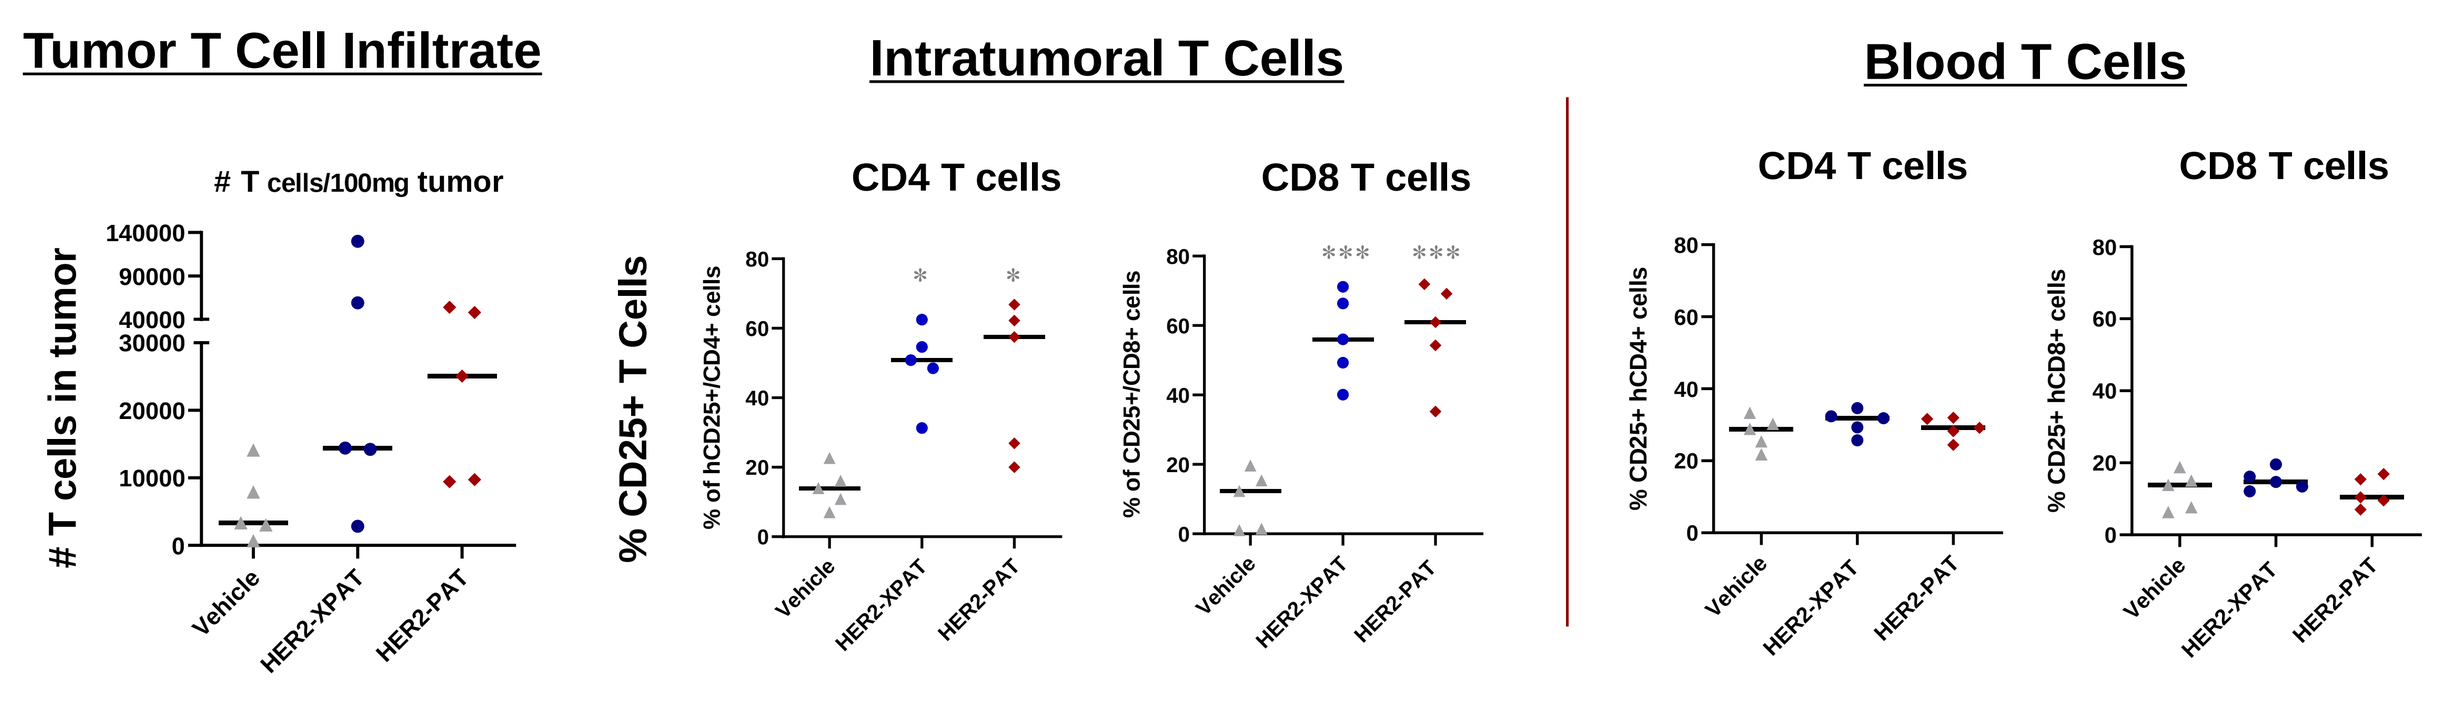
Tumor T Cell Infiltrate
Intratumoral T Cells
Blood T Cells
T cells/100mg tumor
T cells in tumor
% CD25+ T Cells
CD4 T cells
CD8 T cells
Vehicle
HER2-XPAT
HER2-PAT

A) NOG mice were inoculated SC with 2x10^7 BT-474 tumor cells, engrafted with 1x10^7 huPBMCs on Day 8 and treated 2 days later at MTV of 147 mm³ with the indicated test articles at equimolar doses QW for 3 weeks. Lack of tumor growth inhibition by the Non-cleavable HER2-XTEN demonstrates the requirement of protease cleavage for XPAT efficacy **B)** PBMCs were implanted on Day 8 post-BT-474 tumor inoculation, with dosing initiated on Day 21 when tumors averaged 444 mm3. **C)** From an independent BT-474 efficacy experiment conducted as in A), the numbers of tumor infiltrating T cells were quantified per gram of tumor and the activation status of tumor infiltrating and peripheral blood T cells was evaluated by flow cytometry on Day 18 post-TIW dosing of HER2-XPAT and HER2-PAT at 15 nm/kg.

Figure 3. XTEN Masks Significantly Expand Safety Margin of HER2-XPAT vs. PAT in Cynomolgus Monkeys

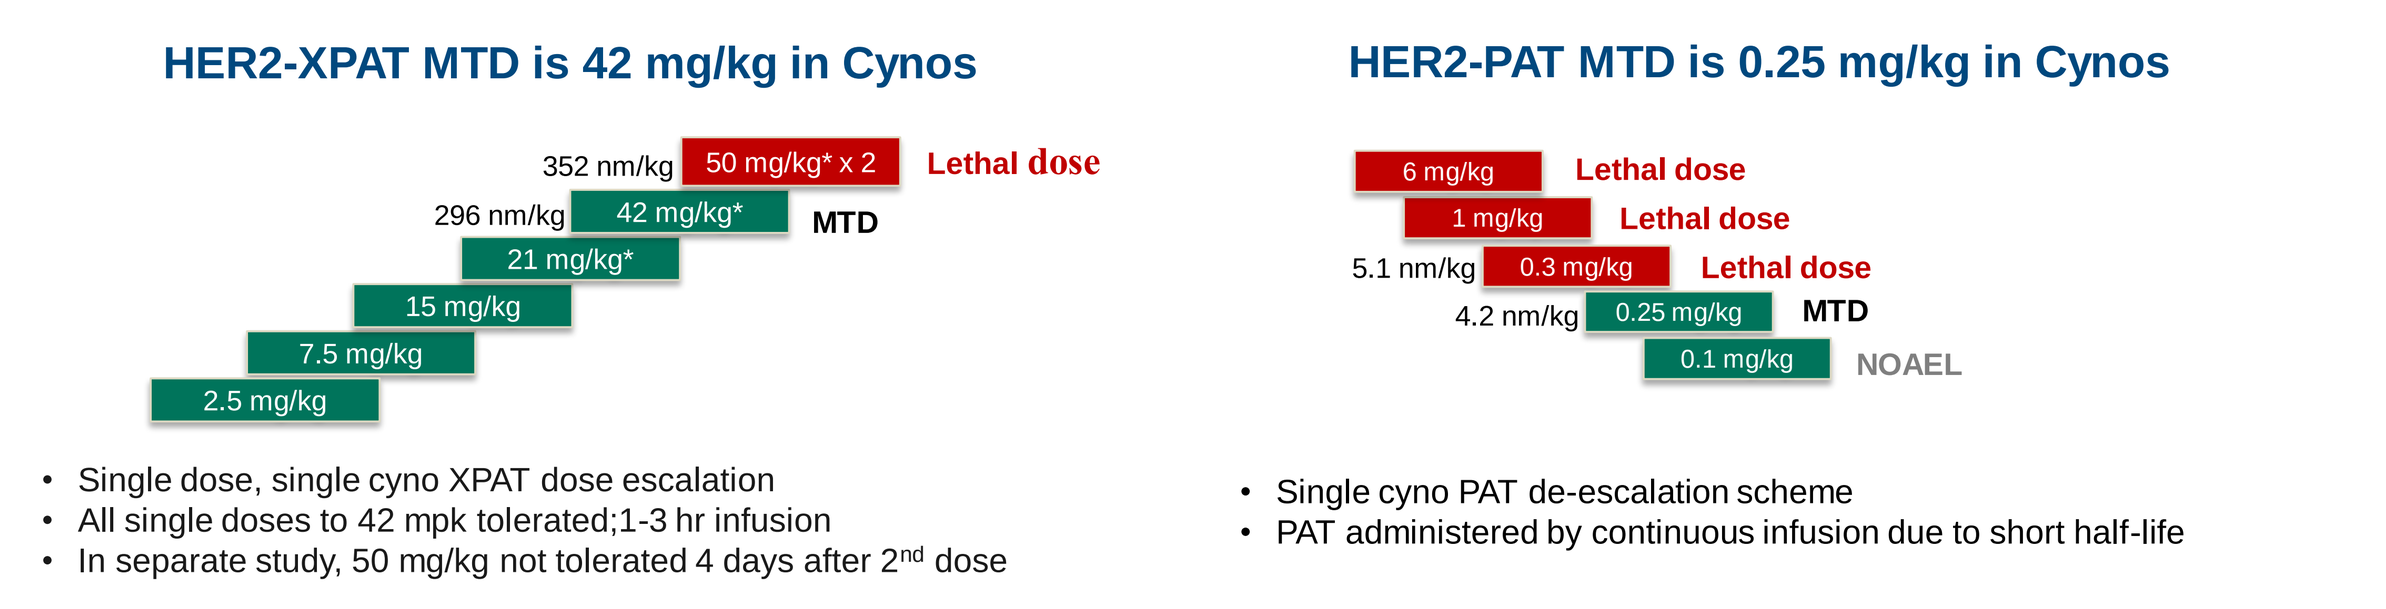
HER2-XPAT MTD is 42 mg/kg in Cynos
352 nm/kg
50 mg/kg* x 2
Lethal dose
296 nm/kg
42 mg/kg*
MTD
21 mg/kg*
15 mg/kg
7.5 mg/kg
2.5 mg/kg
• Single dose, single cyno XPAT dose escalation
• All single doses to 42 mpk tolerated;1-3 hr infusion
• In separate study, 50 mg/kg not tolerated 4 days after 2nd dose
HER2-PAT MTD is 0.25 mg/kg in Cynos
6 mg/kg
Lethal dose
1 mg/kg
Lethal dose
5.1 nm/kg
0.3 mg/kg
Lethal dose
4.2 nm/kg
0.25 mg/kg
MTD
0.1 mg/kg
NOAEL
• Single cyno PAT de-escalation scheme
• PAT administered by continuous infusion due to short half-life

Masked HER2-XPAT Provides ~500-fold Higher Tolerated Cmax vs. Unmasked PAT

Plasma concentrations in Cynomolgus Monkeys

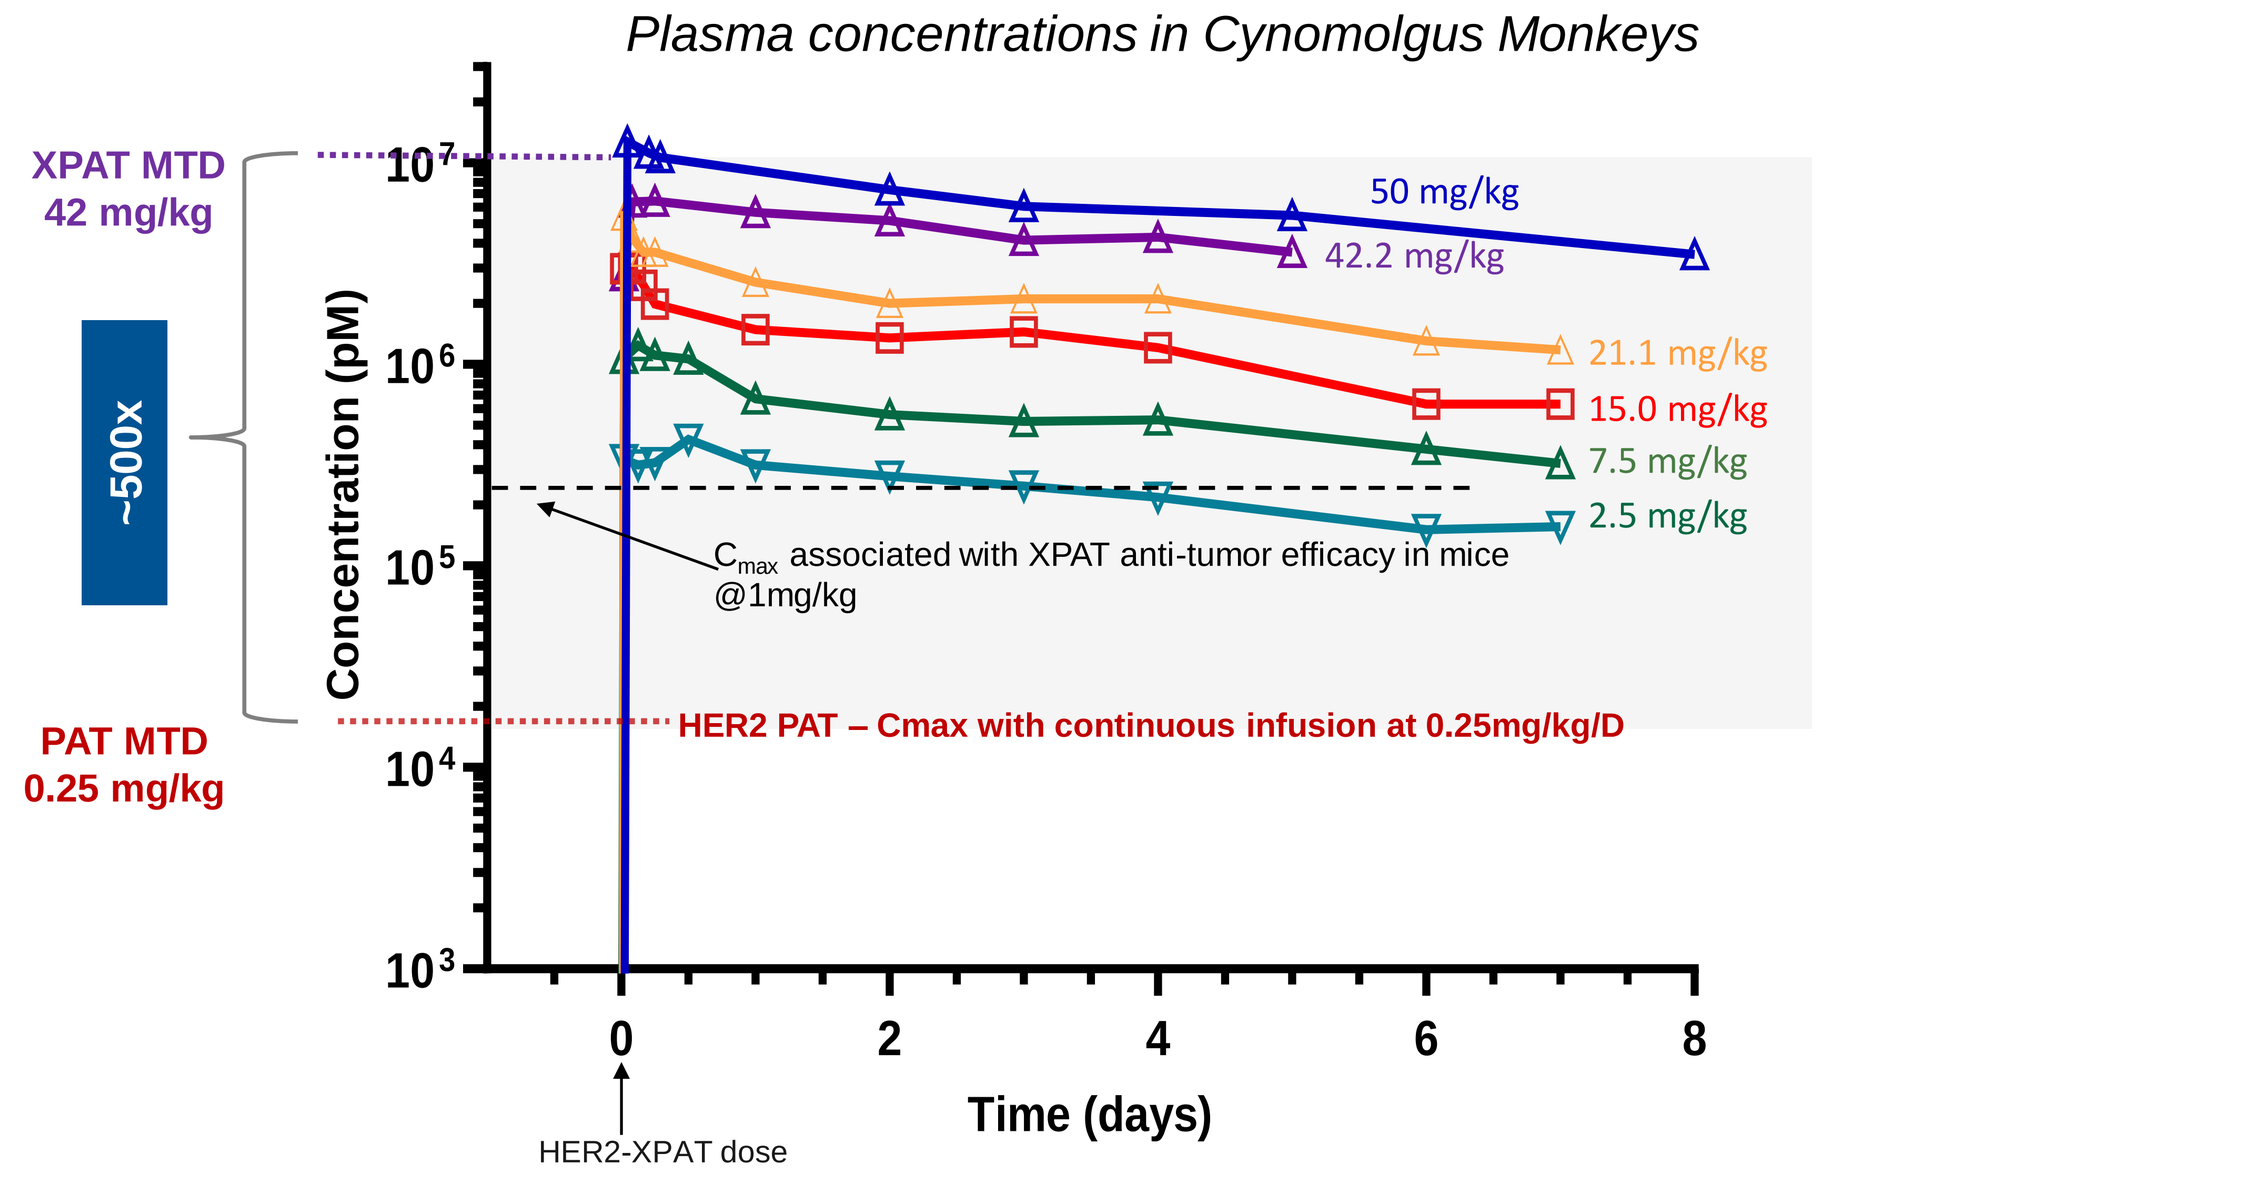
XPAT MTD 42 mg/kg
~500x
PAT MTD 0.25 mg/kg
Concentration (pM)
50 mg/kg
42.2 mg/kg
21.1 mg/kg
15.0 mg/kg
7.5 mg/kg
2.5 mg/kg
Cmax associated with XPAT anti-tumor efficacy in mice @1mg/kg
HER2 PAT – Cmax with continuous infusion at 0.25mg/kg/D
Time (days)
HER2-XPAT dose

HER2-XPAT was administered IV, single dose/animal (doses 2.5-42mpk) and weekly x2 at 50mpk. *At doses 21mpk and above, a variant of HER2-XPAT with a shorter C-terminal XTEN mask was used. HER2-PAT was administered by continuous infusion due to its short half-life. Plasma concentrations of HER2-XPAT were measured by ECLIA* using recombinant HER2 capture and an antibody directed against the XTEN mask for detection. The Cmax values for HER2 PAT were determined by ECLIA utilizing an a-idiotypic Ab directed against the a-CD3 scFv as capture and recombinant HER2 as detection. *ECLIA = Electrochemiluminescent Immunoassay.

Figure 4. HER2-XPAT Induces T Cell Margination at Doses >2.5 mg/kg But Does Not Activate Peripheral T Cells or Induce Cytokine Release Syndrome Even at 50mg/kg

A. T Cell Margination

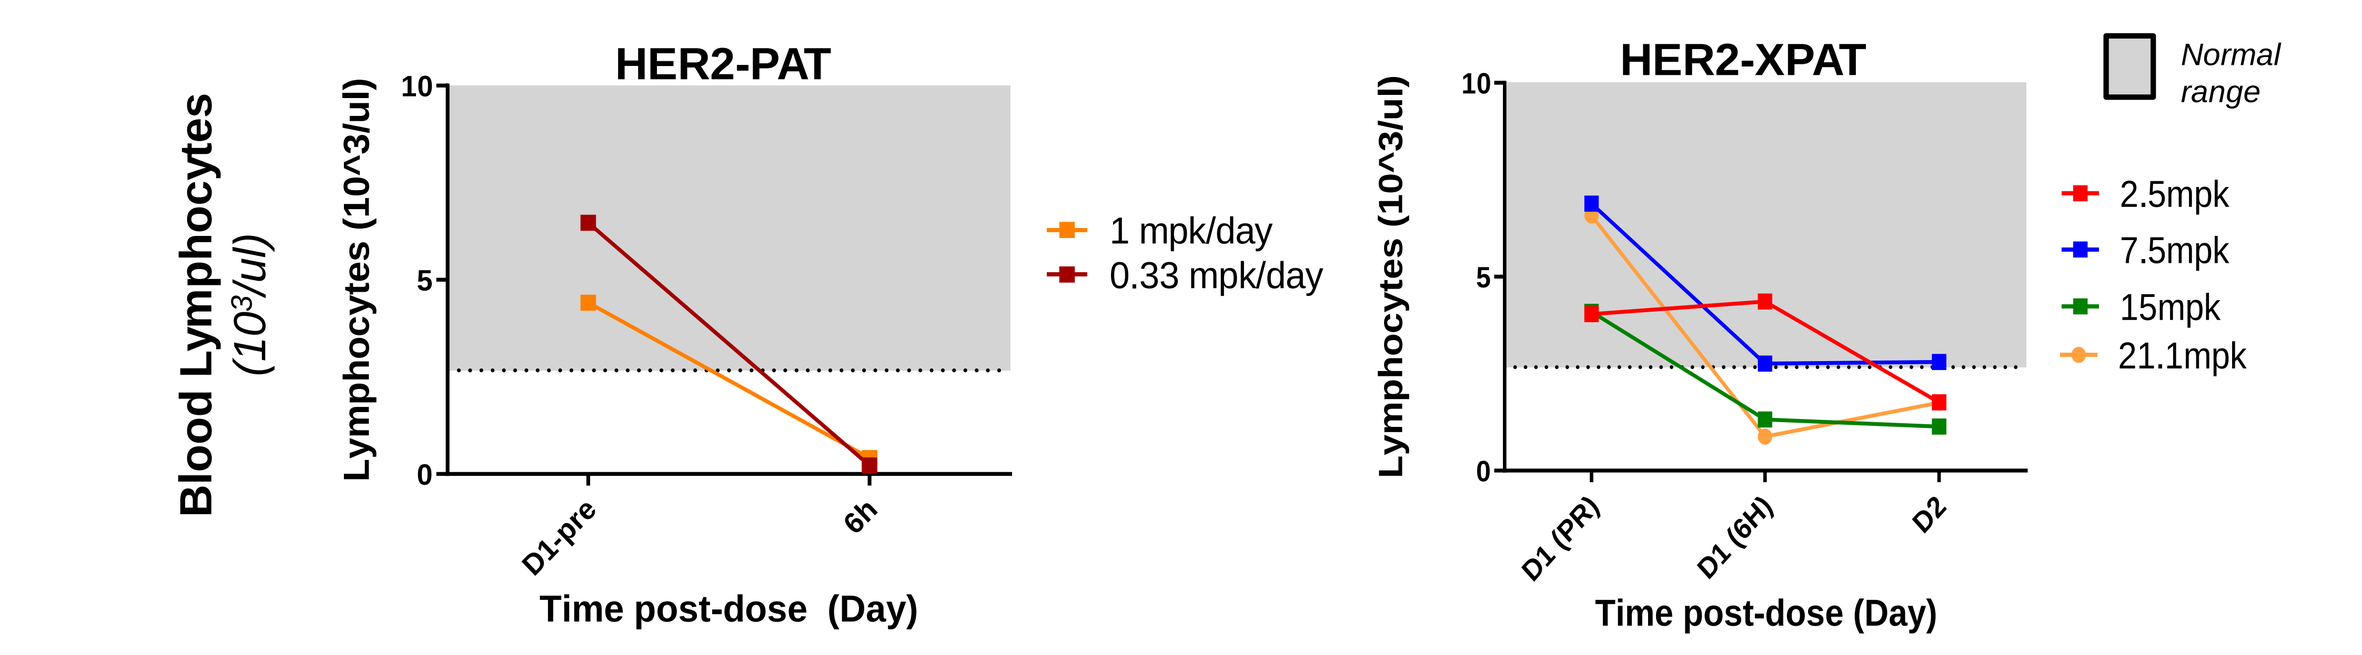
HER2-PAT
HER2-XPAT
Normal range
Blood Lymphocytes (10³/ul)
Lymphocytes (10^3/ul)
1 mpk/day
0.33 mpk/day
2.5mpk
7.5mpk
15mpk
21.1mpk
Time post-dose (Day)

B. Peripheral T Cell Activation

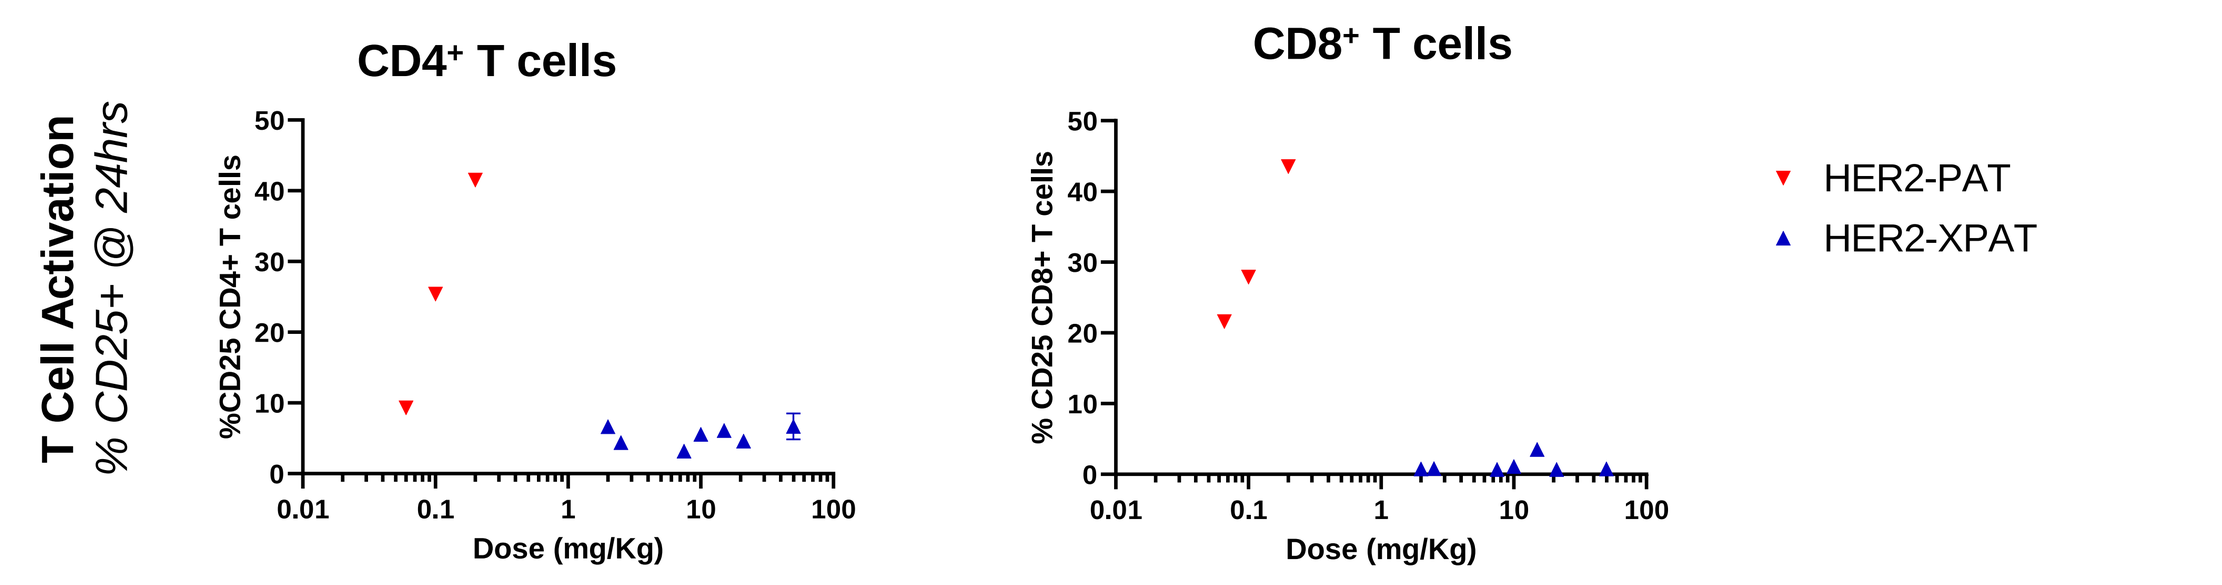
CD4+ T cells
CD8+ T cells
T Cell Activation % CD25+ @ 24hrs
Dose (mg/Kg)
HER2-PAT
HER2-XPAT

C. Plasma Cytokines

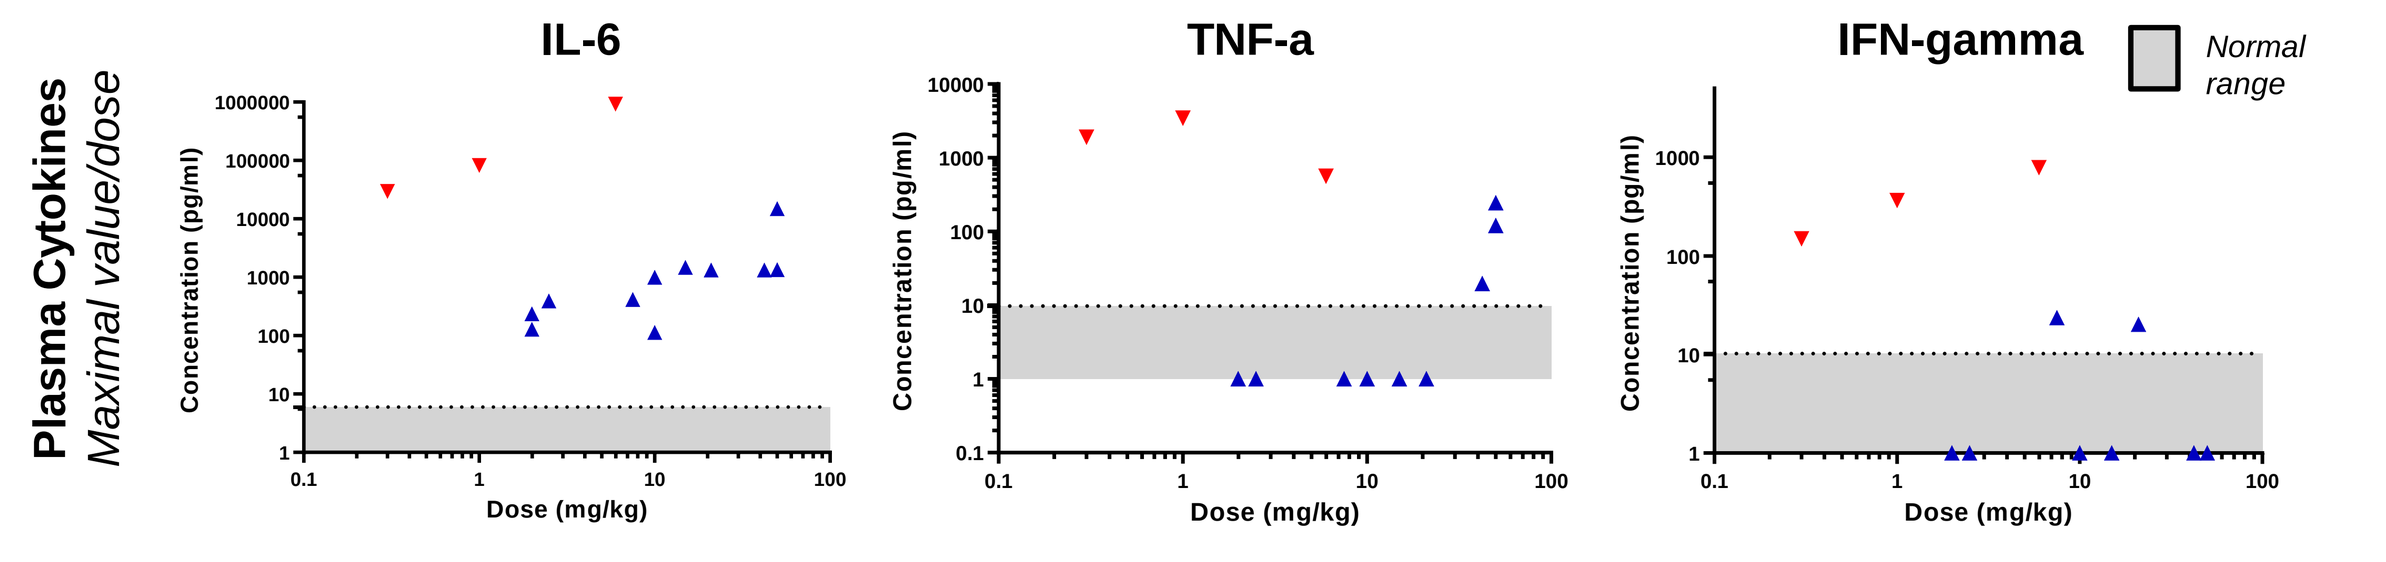
IL-6
TNF-a
IFN-gamma
Normal range
Plasma Cytokines Maximal value/dose
Concentration (pg/ml)
Dose (mg/kg)

A) Blood Lymphocyte counts decline sharply at 6 or 24 hours post-dose, consistent with T cell margination **B)** Peripheral T cell activation (%CD25+) was evaluated by flow cytometry 24 hours post-HER2-XPAT treatment. **C)** Cytokine analysis was performed with a Luminex® suspension array system on plasma samples. Data presented are maximal values measured between 6-24 hours at each evaluated dose.

Figure 5. HER2-XPAT is Largely Stable in Circulation of Cynomolgus Monkeys at 25 mg/kg, Consistent With its Strong Safety Profile

Comparable PK Observed Between HER2-XPAT and Non-Cleavable Format in NHPs

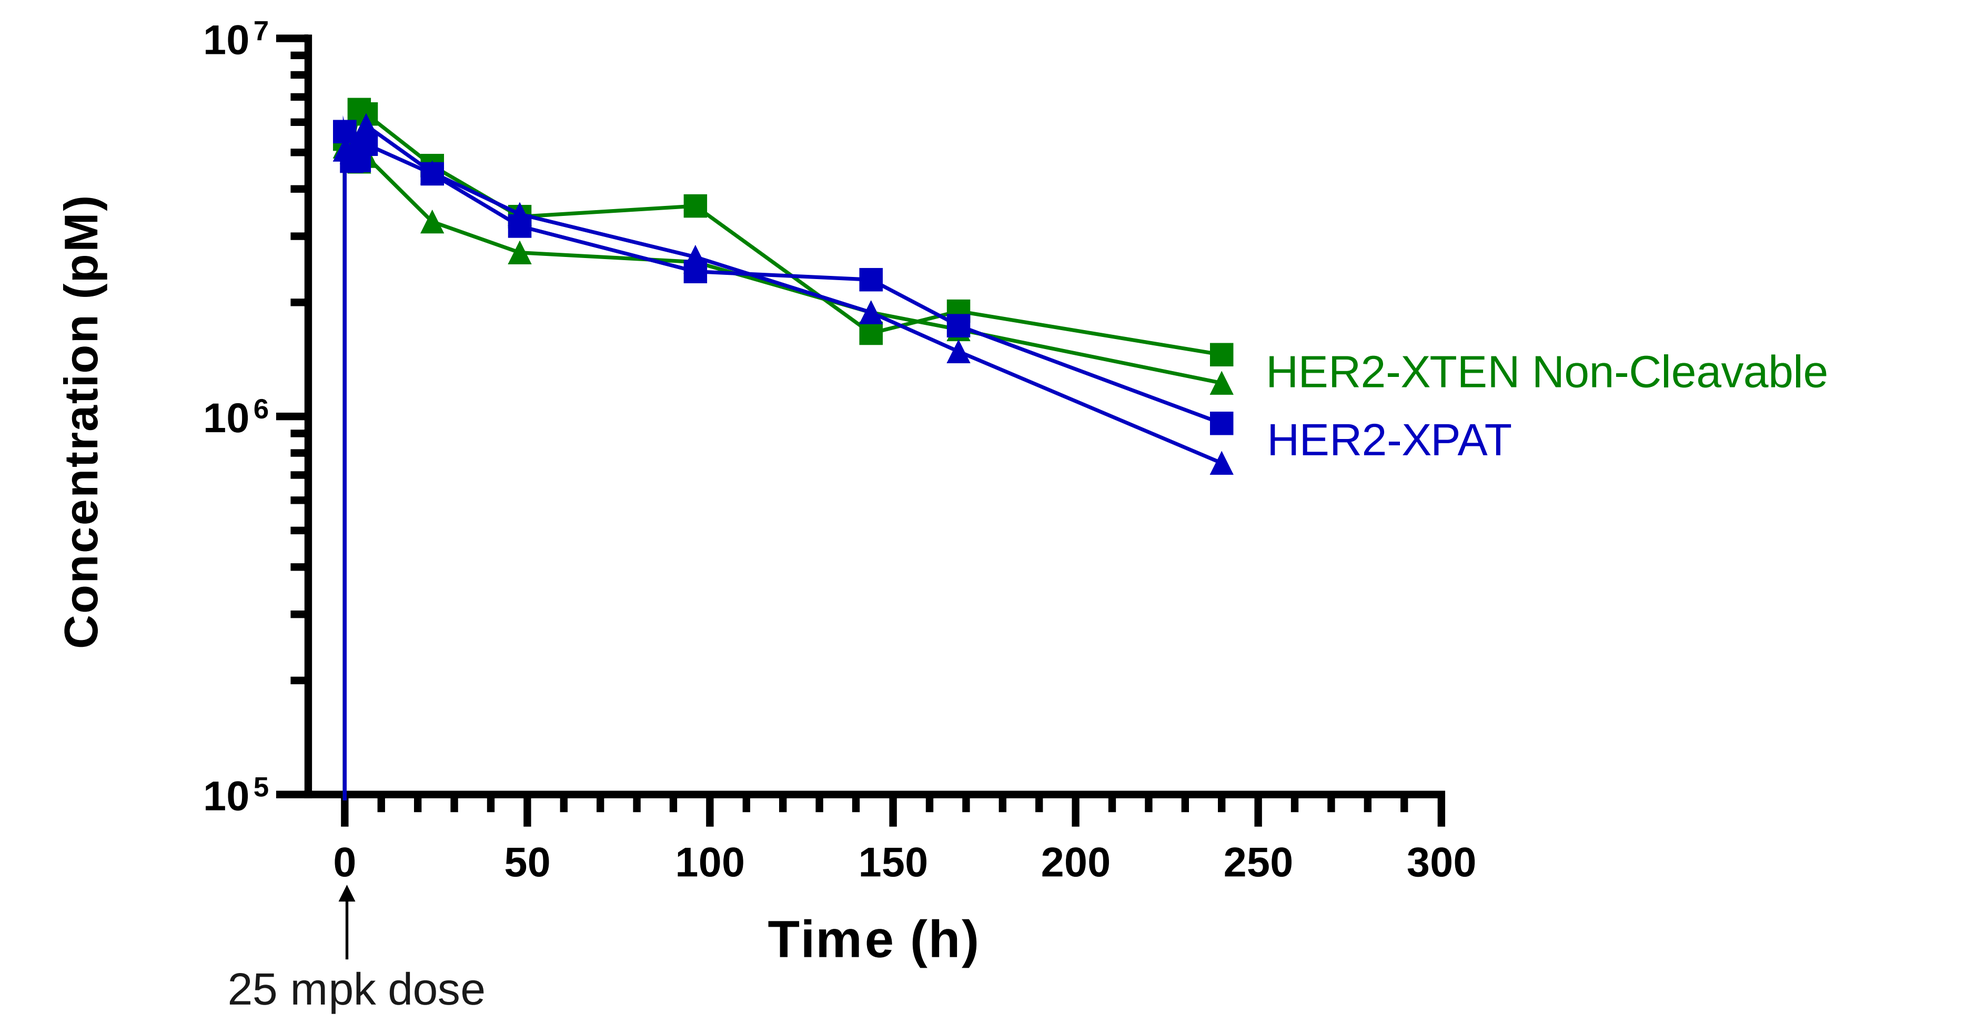
Concentration (pM)
Time (h)
25 mpk dose
HER2-XTEN Non-Cleavable
HER2-XPAT

HER2-XPAT or its non-cleavable counterpart, HER2-XTEN were administered as a single IV dose of 25 mg/kg. Plasma drug concentrations were measured by ECLIA* using recombinant HER2 as capture and an antibody directed against the XTEN mask for detection. *ECLIA = Electrochemiluminescent Immunoassay

Figure 6. Negligible Amounts of Fully Active PAT are Generated *in vitro* in Plasma Samples from Patients with Cancer and Inflammatory Diseases

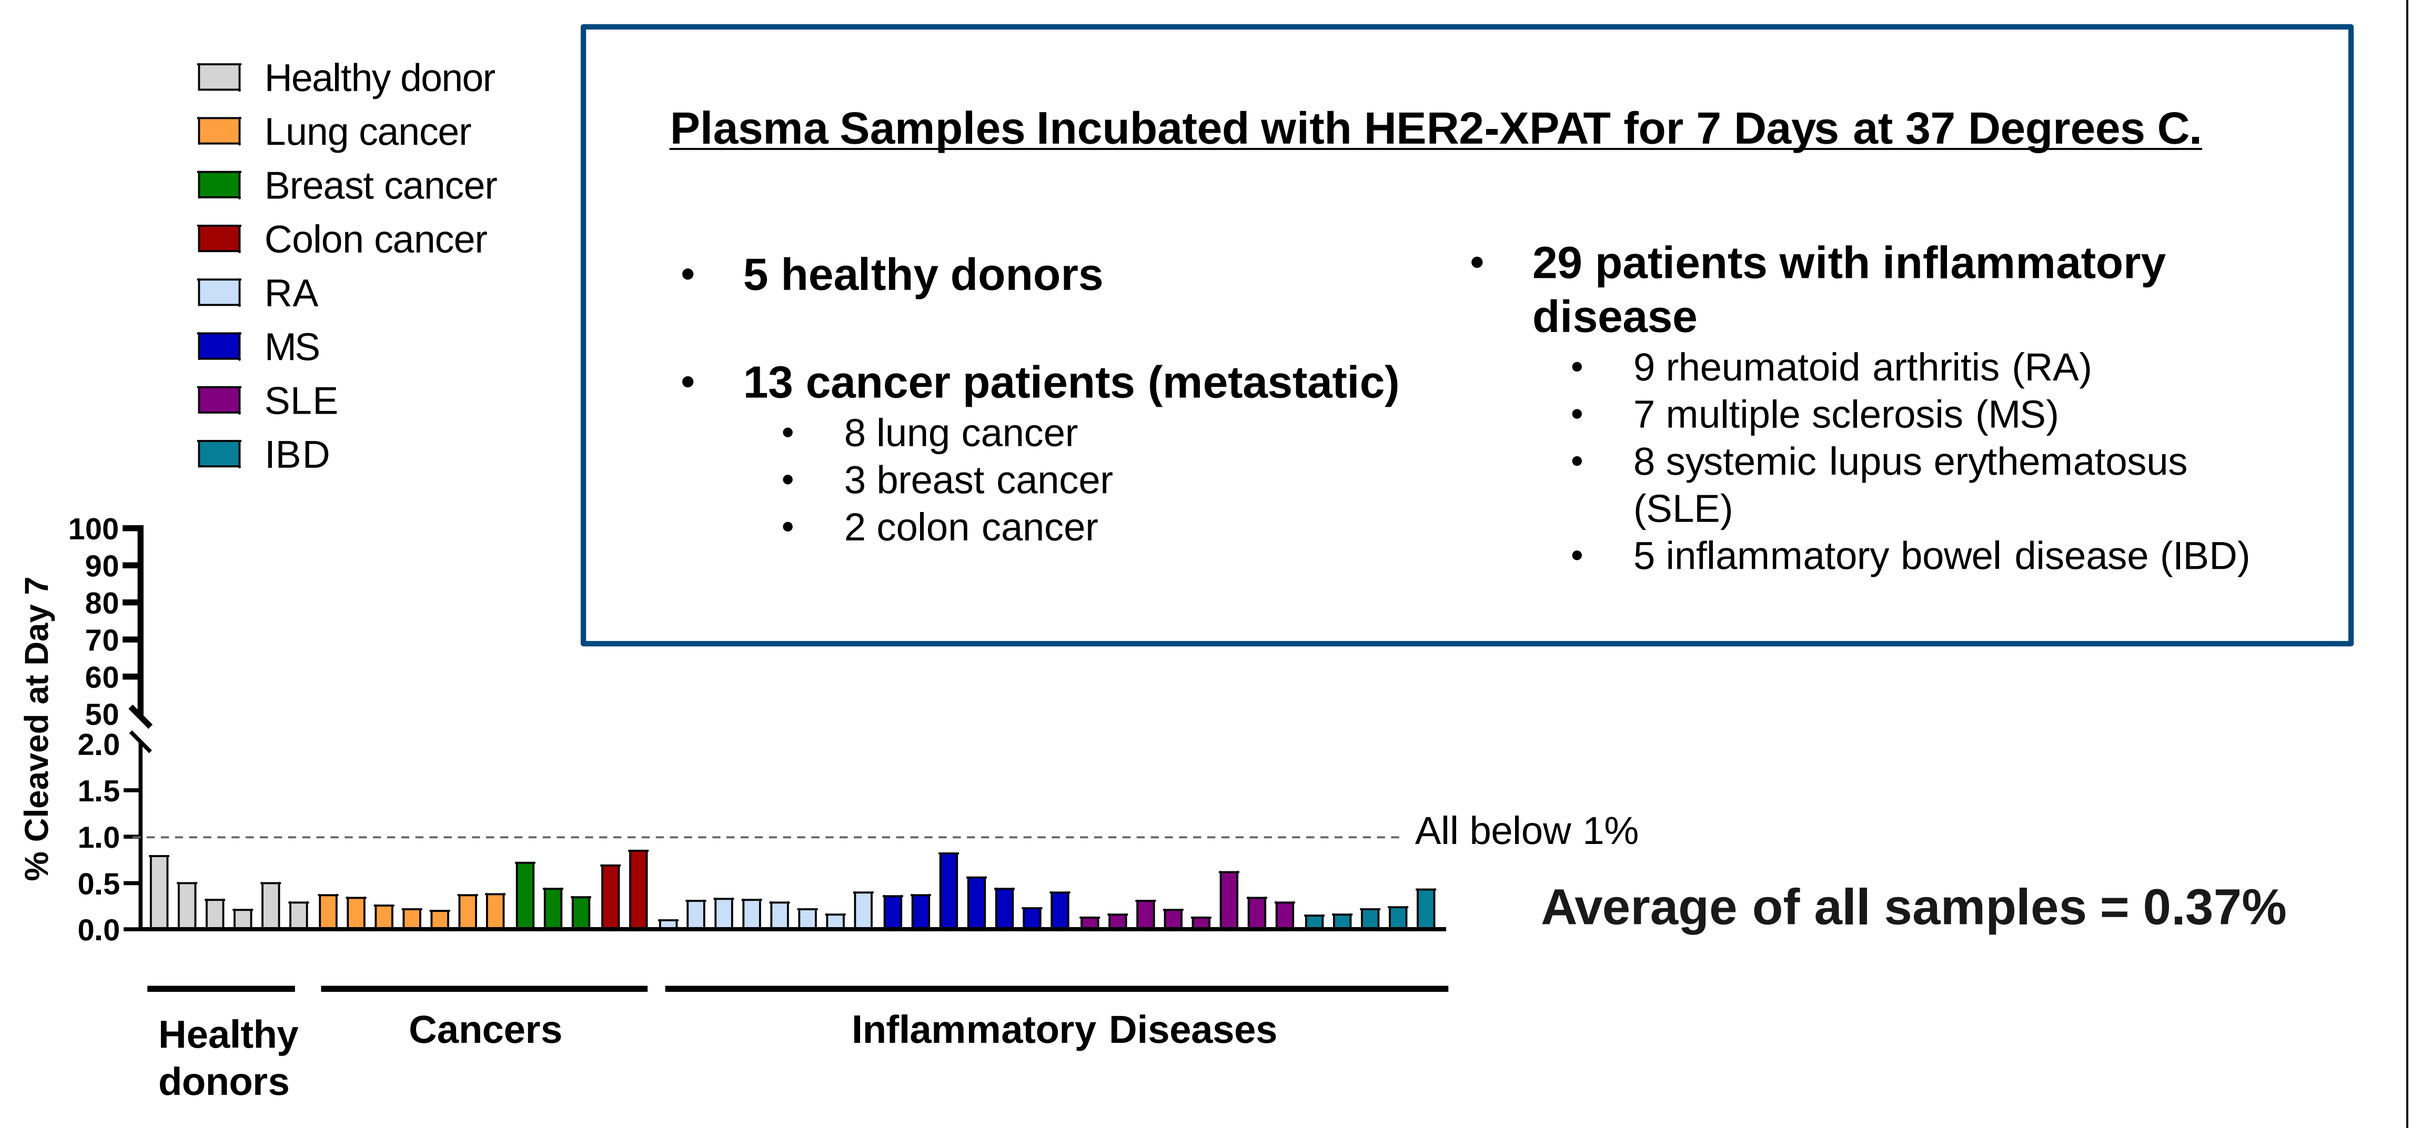
Healthy donor
Lung cancer
Breast cancer
Colon cancer
RA
MS
SLE
IBD
Plasma Samples Incubated with HER2-XPAT for 7 Days at 37 Degrees C.
• 5 healthy donors
• 13 cancer patients (metastatic)
• 8 lung cancer
• 3 breast cancer
• 2 colon cancer
• 29 patients with inflammatory disease
• 9 rheumatoid arthritis (RA)
• 7 multiple sclerosis (MS)
• 8 systemic lupus erythematosus (SLE)
• 5 inflammatory bowel disease (IBD)
% Cleaved at Day 7
All below 1%
Average of all samples = 0.37%
Healthy donors
Cancers
Inflammatory Diseases

A) A DyLight-labeled HER2-XPAT molecule was incubated for 7 days at 37 degrees C in plasma from healthy human donors or from patients with the indicated cancers or systemic autoimmune diseases. The degree of PAT generation was determined by size exclusion chromatography exploiting the fluorescence from the DyLight-labeling to increase sensitivity of detection.

SUMMARY/CONCLUSIONS

- *In vitro*, proteolytically-unmasked HER2-XPATs demonstrate potent cytotoxicity against tumor lines with EC50s in the single-digit pM range. XTEN masking reduces target-directed T cell cytotoxicity and T cell activation by up to 13,000-fold
- In the established BT-474 xenograft model, HER2-XPAT induced protease-dependent tumor regressions at equimolar doses as the unmasked (active) T cell engager
- In cynomolgus monkeys, HER2-XPAT demonstrated a high safety margin, supported by its protease stability in circulation and a maximum tolerated exposure that was ~500 fold higher than that of its active form (PAT). No CRS or systemic T cell activation was observed even at 50 mg/kg supportive of minimal CRS risk for XPATs vs standard TCEs
- Negligible cleavage of XPAT to fully active PAT occurred in vitro following extended incubation at 37 degrees C in plasma from patients with cancer or systemic inflammatory disorders, consistent with the abundance of protease inhibitors in circulation even in disease states
- XPATs represent a novel strategy to improve the toxicity profile of T cell engagers while maintaining their potency against solid tumors, thus enabling a significant increase in the therapeutic index and expansion of target landscape for this potent modality